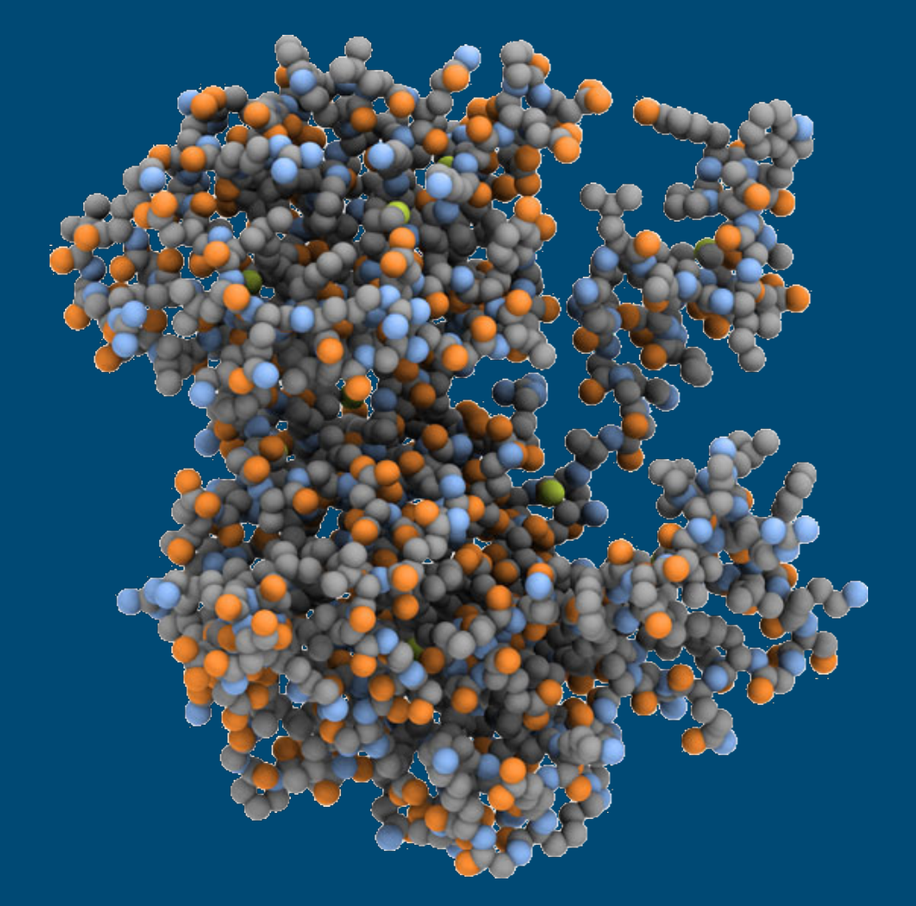

NDI-101150 Is a Potent and Highly Selective Hematopoietic Progenitor Kinase 1 (HPK1) Inhibitor That Promotes a Robust and Broad Anti-Tumor Immune Response

David Ciccone[a], Fu-Shan Kuo, Scott Boiko, Scott Daigle, Samantha Carreiro, Neelu Kaila, Gene Yau, Esha A. Gangolli, Denise Levasseur, Bhaskar Srivastava, Frank G. Basile, and Christine Loh

Nimbus Therapeutics, Boston, MA, USA

nimbus THERAPEUTICS

BACKGROUND

- Hematopoietic progenitor kinase 1 (HPK1) is a member of the MAP4K family of protein serine/threonine kinases that negatively regulates activation signals in multiple immune cells and is an attractive therapeutic target in many cancers (**Figure 1**).[1,2]
- Using structure-based drug design, we developed a highly selective HPK1 inhibitor, NDI-101150, with nanomolar potency and physiochemical properties suitable for once daily oral administration.
- NDI-101150 enhanced the activity of B cells ex vivo, increased antigen-specific antibody production in vivo, and induced robust tumor growth inhibition in an EMT-6 syngeneic model.[3]
- Preliminary clinical data indicate that NDI-101150 has antitumor activity and good tolerability in patients with advanced solid tumors.[4] We report the effects of NDI-101150 on a range of lymphoid and myeloid cells, as well as its antitumor and pharmacodynamic immunomodulatory activity.

Figure 1: HPK1 is a Compelling Immuno-Oncology Therapeutic Target

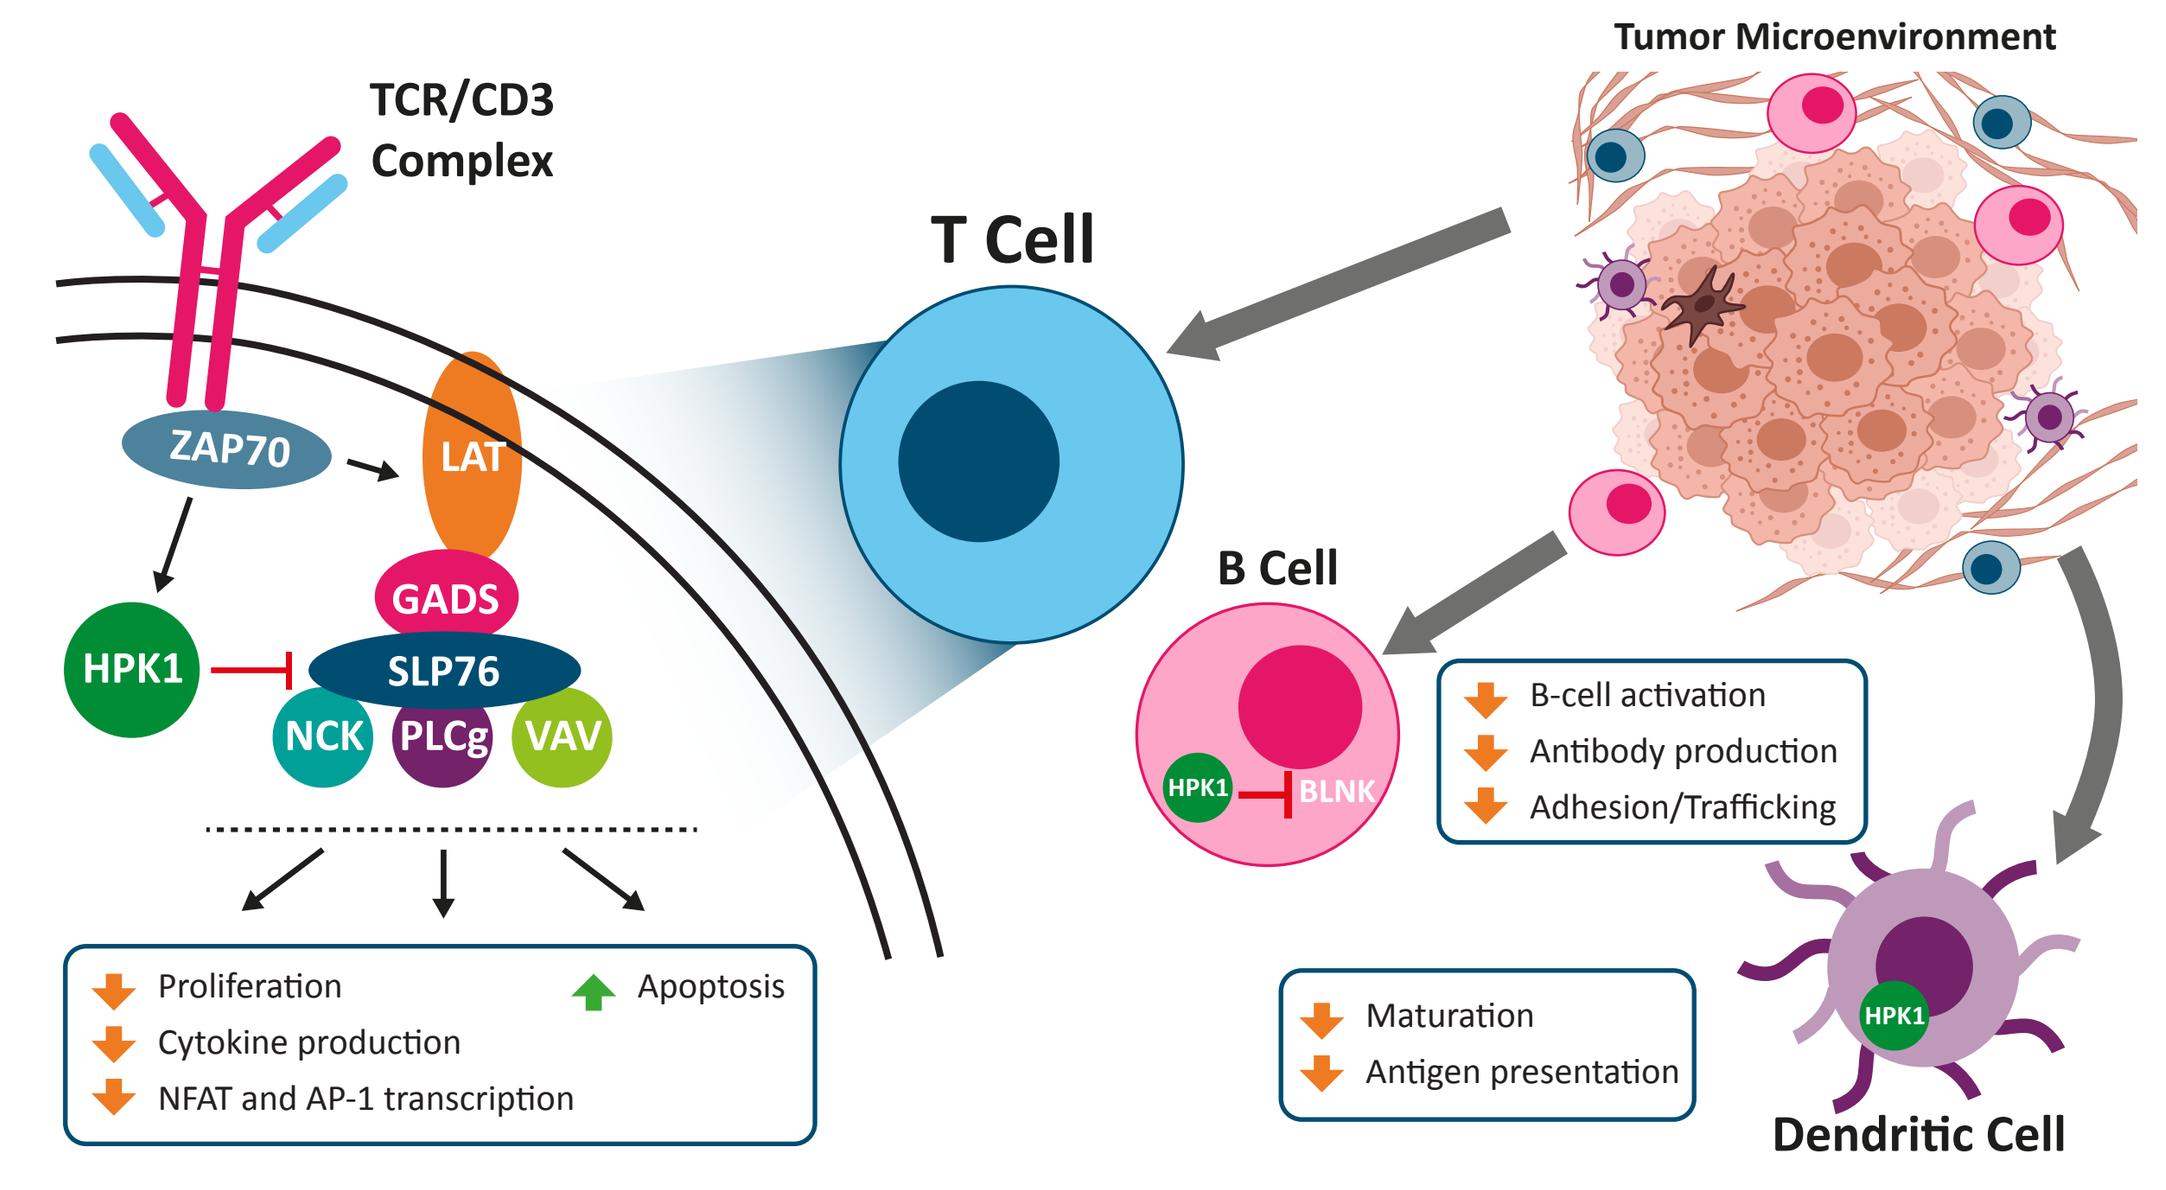
TCR/CD3 Complex
ZAP70
LAT
GADS
HPK1
SLP76
NCK
PLCg
VAV
Proliferation
Apoptosis
Cytokine production
NFAT and AP-1 transcription
T Cell
Tumor Microenvironment
B Cell
BLNK
B-cell activation
Antibody production
Adhesion/Trafficking
Maturation
Antigen presentation
Dendritic Cell

AP-1, activator protein 1; BLNK, B-cell linker protein; GADS, GRB2 related adaptor protein downstream of Shc; HPK1, hematopoietic progenitor kinase 1; LAT, linker for activation of T cells; NCK, non-catalytic region of the tyrosine kinase; NFAT, nuclear factor of activated T cells; PLCg, phospholipase C, gamma 1; SLP76, SH2 domain containing leukocyte protein of 76kDa; TCR, T cell receptor; ZAP70, zeta-chain-associated protein kinase 70

- Negative regulator of T cell-, B cell-, and dendritic cell-mediated immune responses
- Tissue-specific expression – only in hematopoietic cells
- Genetically validated target:
 - HPK1–/– mice are resistant to growth of Lewis lung carcinoma and have enhanced antitumor T cell responses
 - HPK1 kinase-inactive knock-in mice show impaired GL261 tumor growth, which is associated with increased T cell infiltration

METHODS/RESULTS

Figure 2. NDI-101150 is a Potent and Exquisitely Selective HPK1 Inhibitor

A structure-based drug design approach was used to generate potent and selective inhibitors of HPK1. Biochemical and biophysical assays, as well as primary human and mouse immune cell-based activation assays, were utilized for multiple rounds of structure-activity relationship (SAR) studies. NDI-101150 was identified as a highly potent HPK1 inhibitor that shows high selectivity against T cell-specific kinases and kinases in the MAP4K family.

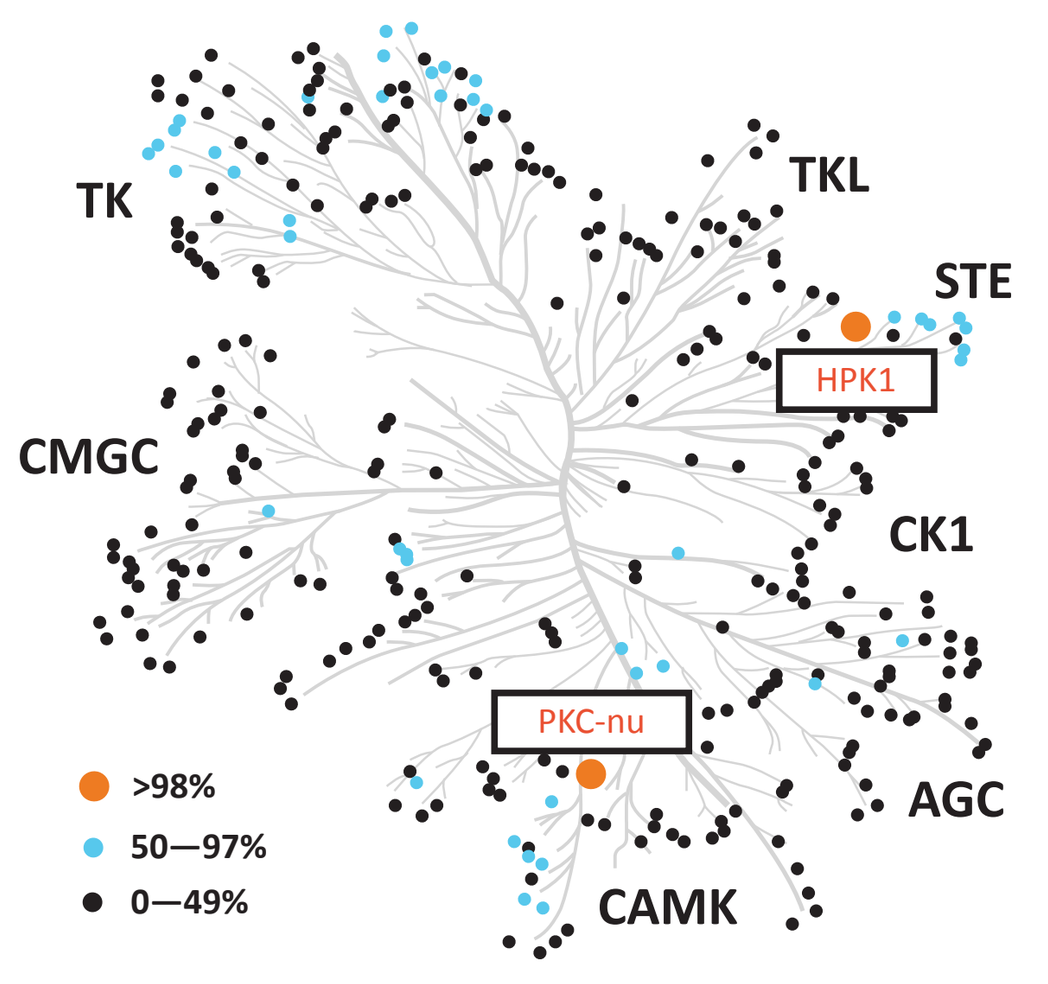
TK
TKL
STE
HPK1
CMGC
CK1
PKC-nu
AGC
CAMK
>98%
50—97%
0—49%

NDI-101150	
Potency	
HPK1 biochemical IC_{50}	0.7 nM
HPK1 cellular IC_{50}	41 nM
FOLD selectivity against MAP4K family	
GLK (@ 1 mM ATP)	377
KHS (@ 1 mM ATP)	489
TNIK (@ 1 mM ATP)	1,336
HGK (@ 1 mM ATP)	>10,000
MINK (@ 1 mM ATP)	>10,000
FOLD selectivity against immune cell kinases	
FYN (@ 1 mM ATP)	3,110
c-SRC (@ 1 mM ATP)	3,630
LCK (@ 1 mM ATP)	2,143
GCK (@ 10 µM ATP)	>8,000
SYK (@ 10 µM ATP)	>20,000

Figure 3. NDI-101150 Inhibits HPK1-Mediated Phosphorylation of SLP76 and BLNK in Purified Human Lymphocytes

T Cells: PBMCs were isolated from human donors and treated with NDI-101150 or DMSO for 1 hour before stimulating with anti-CD3/28 for 30 minutes. Phospho-SLP76 (S376) was detected by FACS in both CD4+ and CD8+ T cells.

B Cells: CD19+ B cells were purified from human donors and allowed to rest overnight. The following day, cells were treated with NDI-101150 or DMSO for 1 hour before stimulating with IL-21, IL-4, and anti-CD40 antibody for 30 minutes. Whole cell extracts were prepared and phospho-BLNK (Thr 152) was detected by western blot.

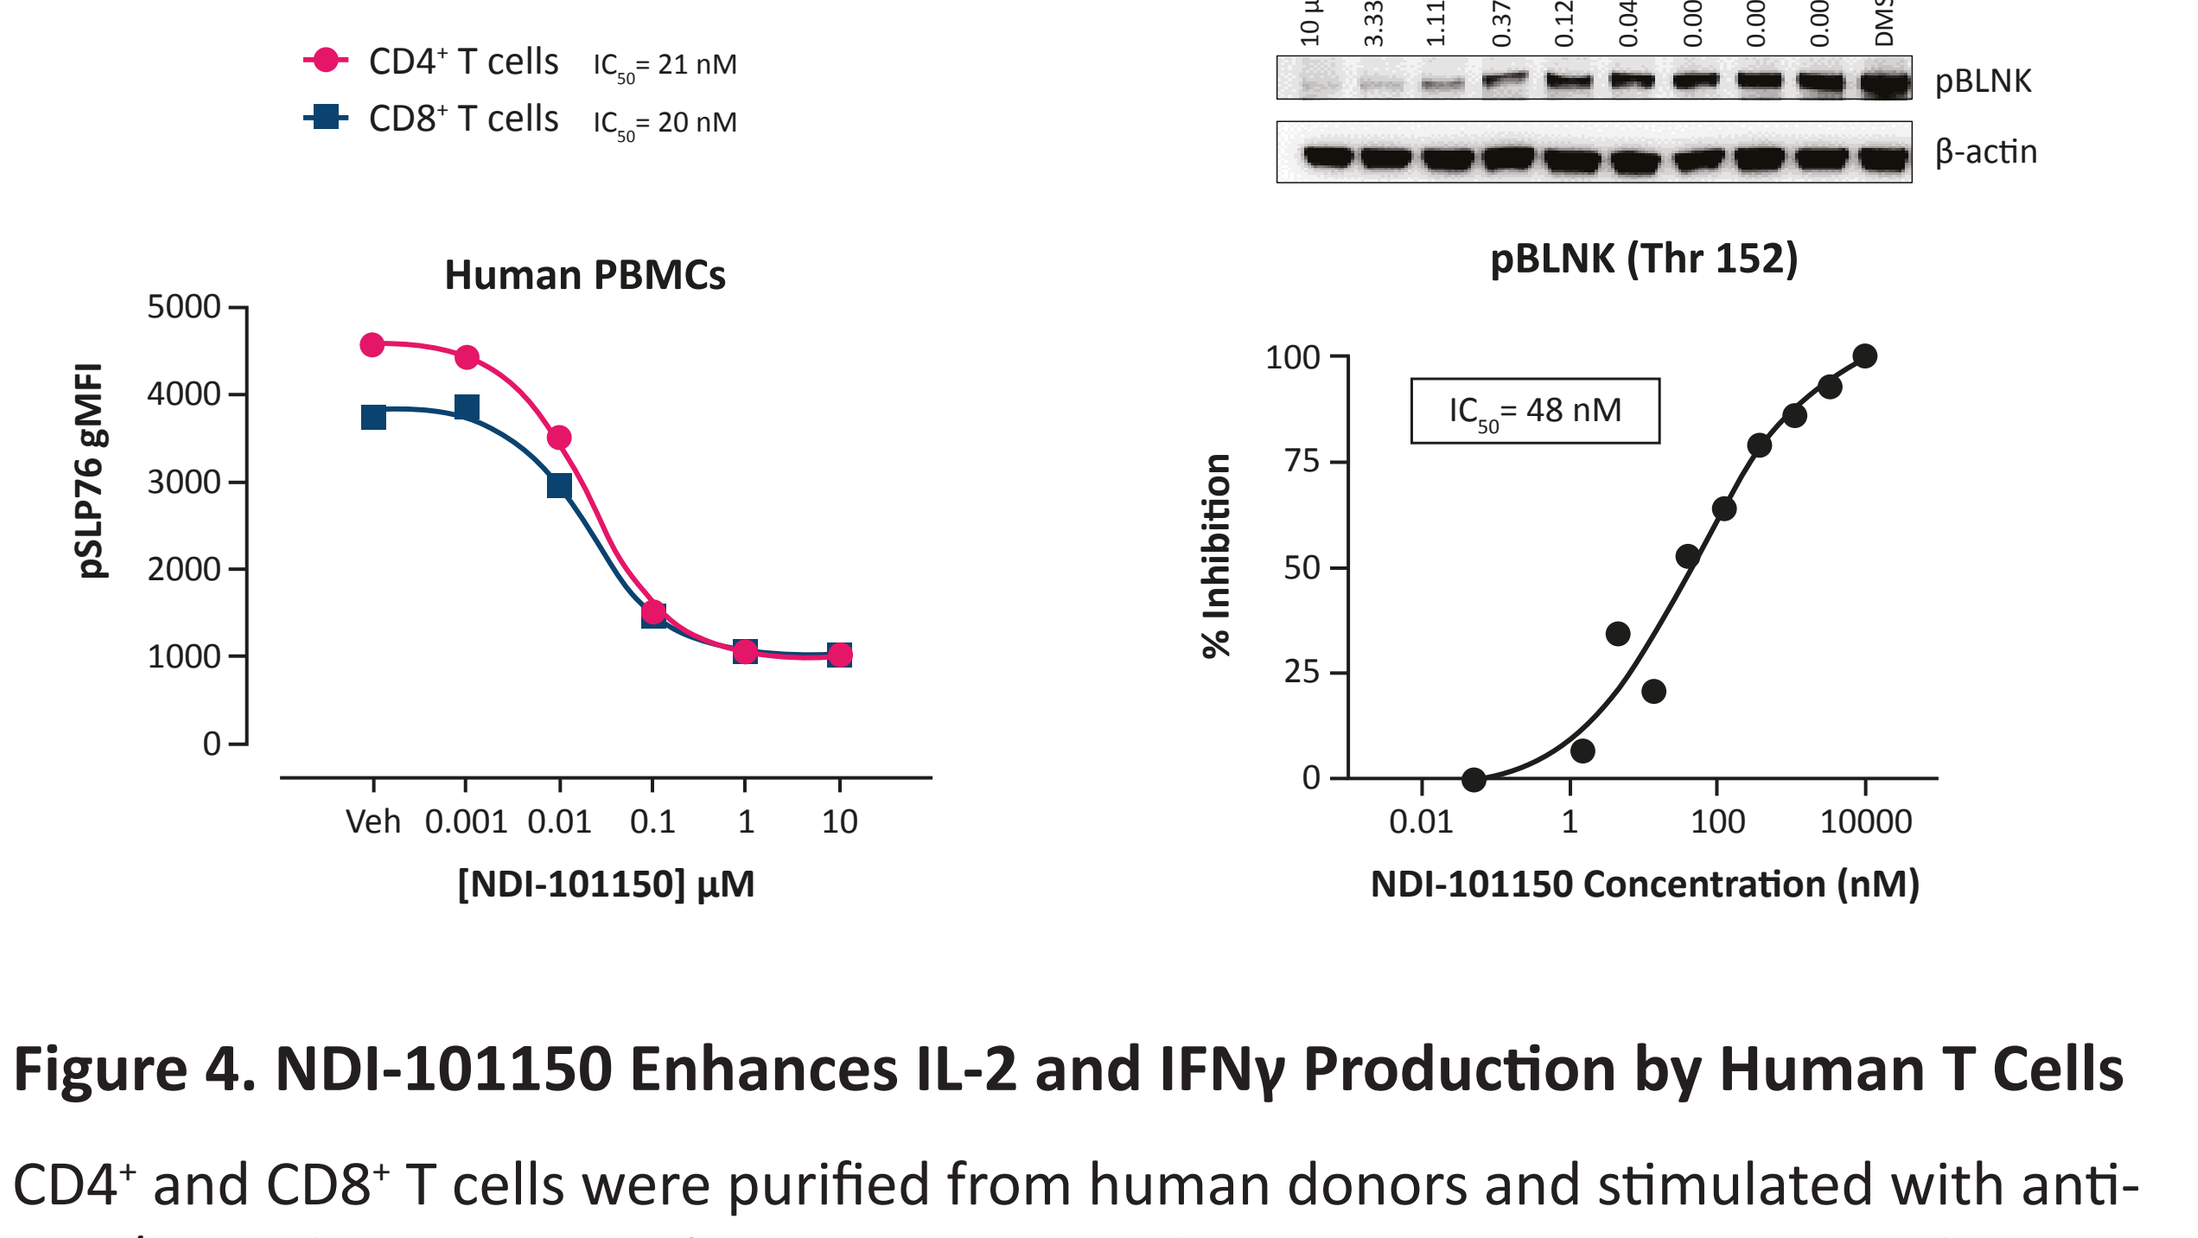
CD4+ T cells IC_{50} = 21 nM
CD8+ T cells IC_{50} = 20 nM
Human PBMCs
pSLP76 gMFI
[NDI-101150] µM
10 µM
3.33 µM
1.11 µM
0.370 µM
0.123 µM
0.0412 µM
0.0037 µM
0.0046 µM
0.0015 µM
DMSO
pBLNK
β-actin
pBLNK (Thr 152)
IC_{50} = 48 nM
% Inhibition
NDI-101150 Concentration (nM)

Figure 4. NDI-101150 Enhances IL-2 and IFNγ Production by Human T Cells

CD4+ and CD8+ T cells were purified from human donors and stimulated with anti-CD3/28 in the presence of NDI-101150. Cytokine production was assayed by MSD.

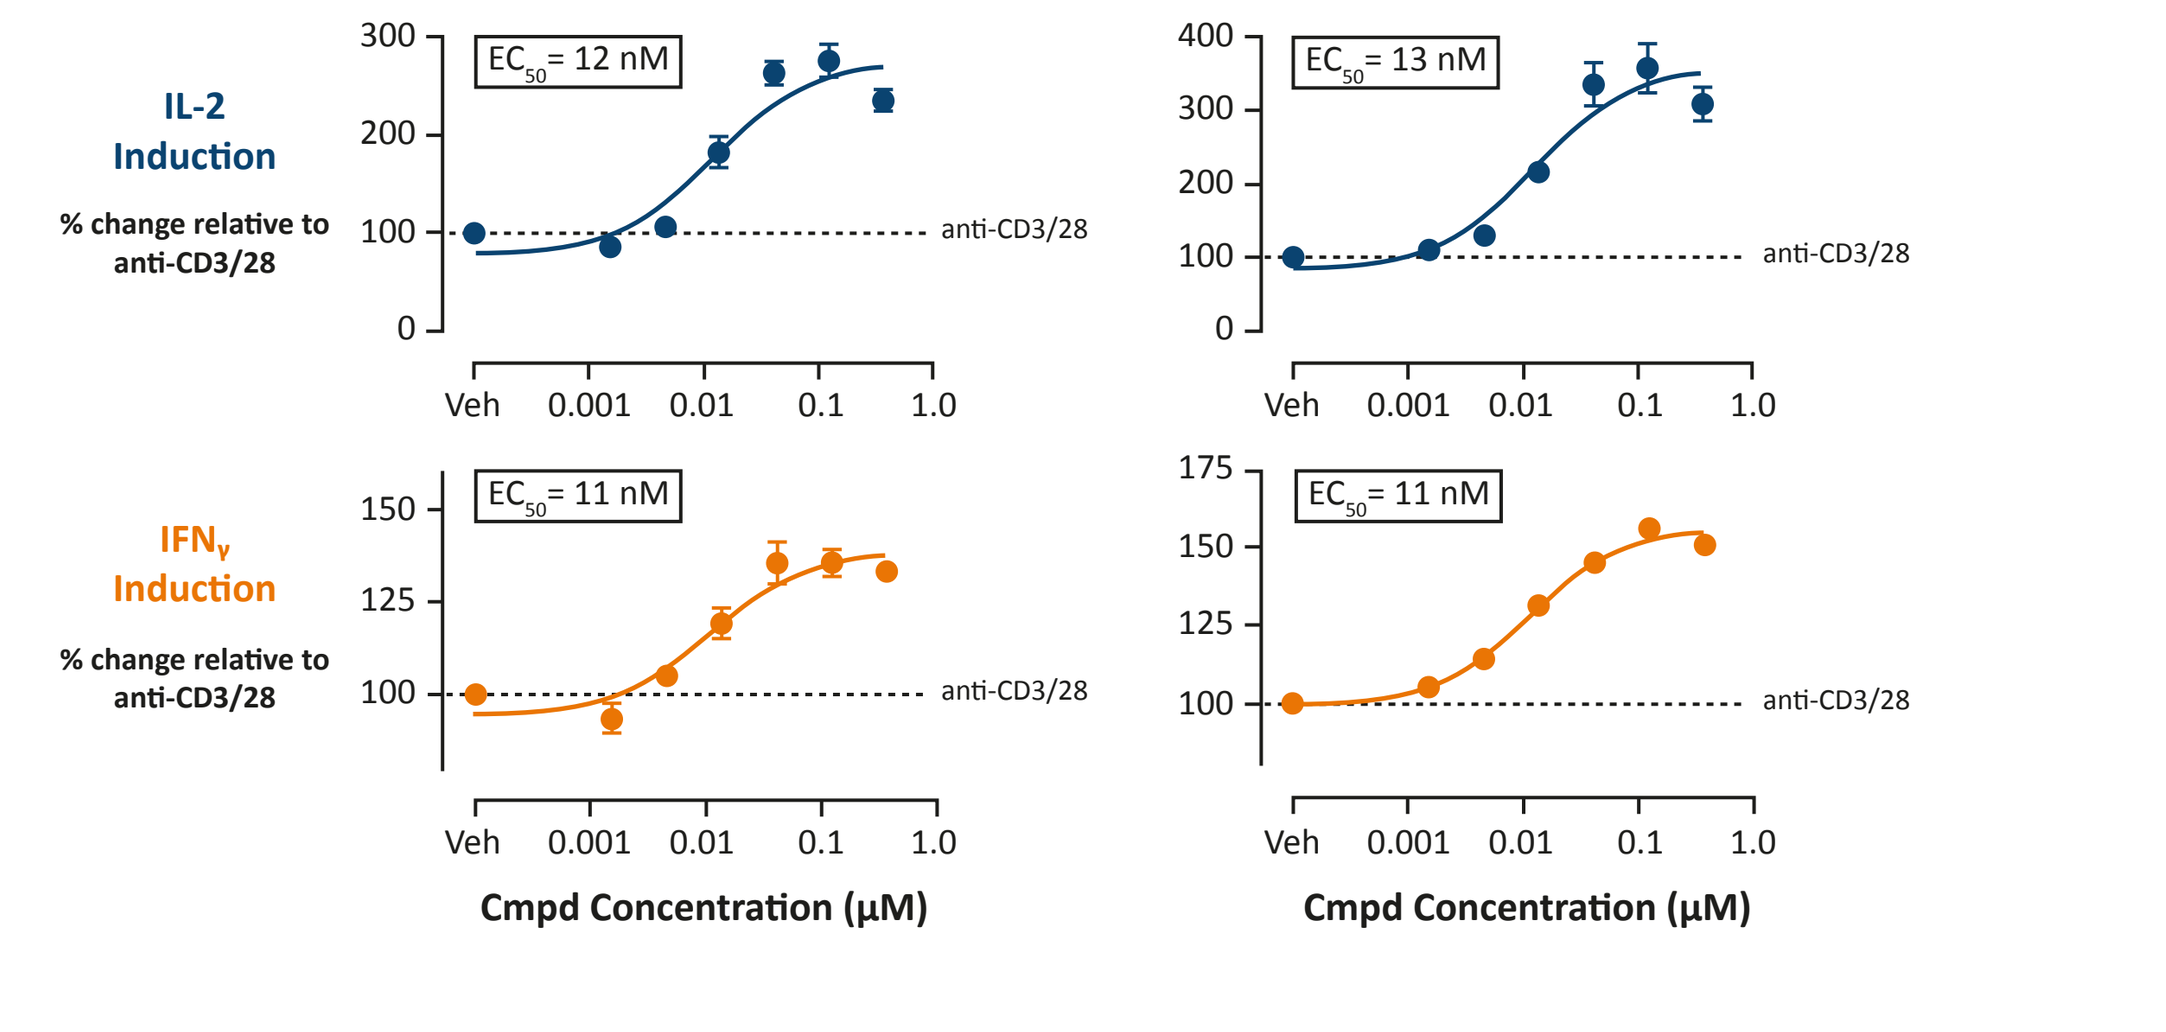
CD4+ T cells
CD8+ T cells
IL-2 Induction
% change relative to anti-CD3/28
EC_{50} = 12 nM
EC_{50} = 13 nM
IFNγ Induction
EC_{50} = 11 nM
EC_{50} = 11 nM
anti-CD3/28
Cmpd Concentration (µM)

METHODS/RESULTS

Figure 5. NDI-101150 Reinvigorates Exhausted Human T Cells and Demonstrates Synergy with anti-PD1

T cells purified from human donors were exhausted in vitro through 5 iterative cycles of stimulation with anti-CD3/28 for 24 hrs followed by resting for 24 hrs. Exhausted T cells were then used in MLR experiments in the presence of NDI-101150, anti-PD1, or both, and cytokine production was measured by MSD.

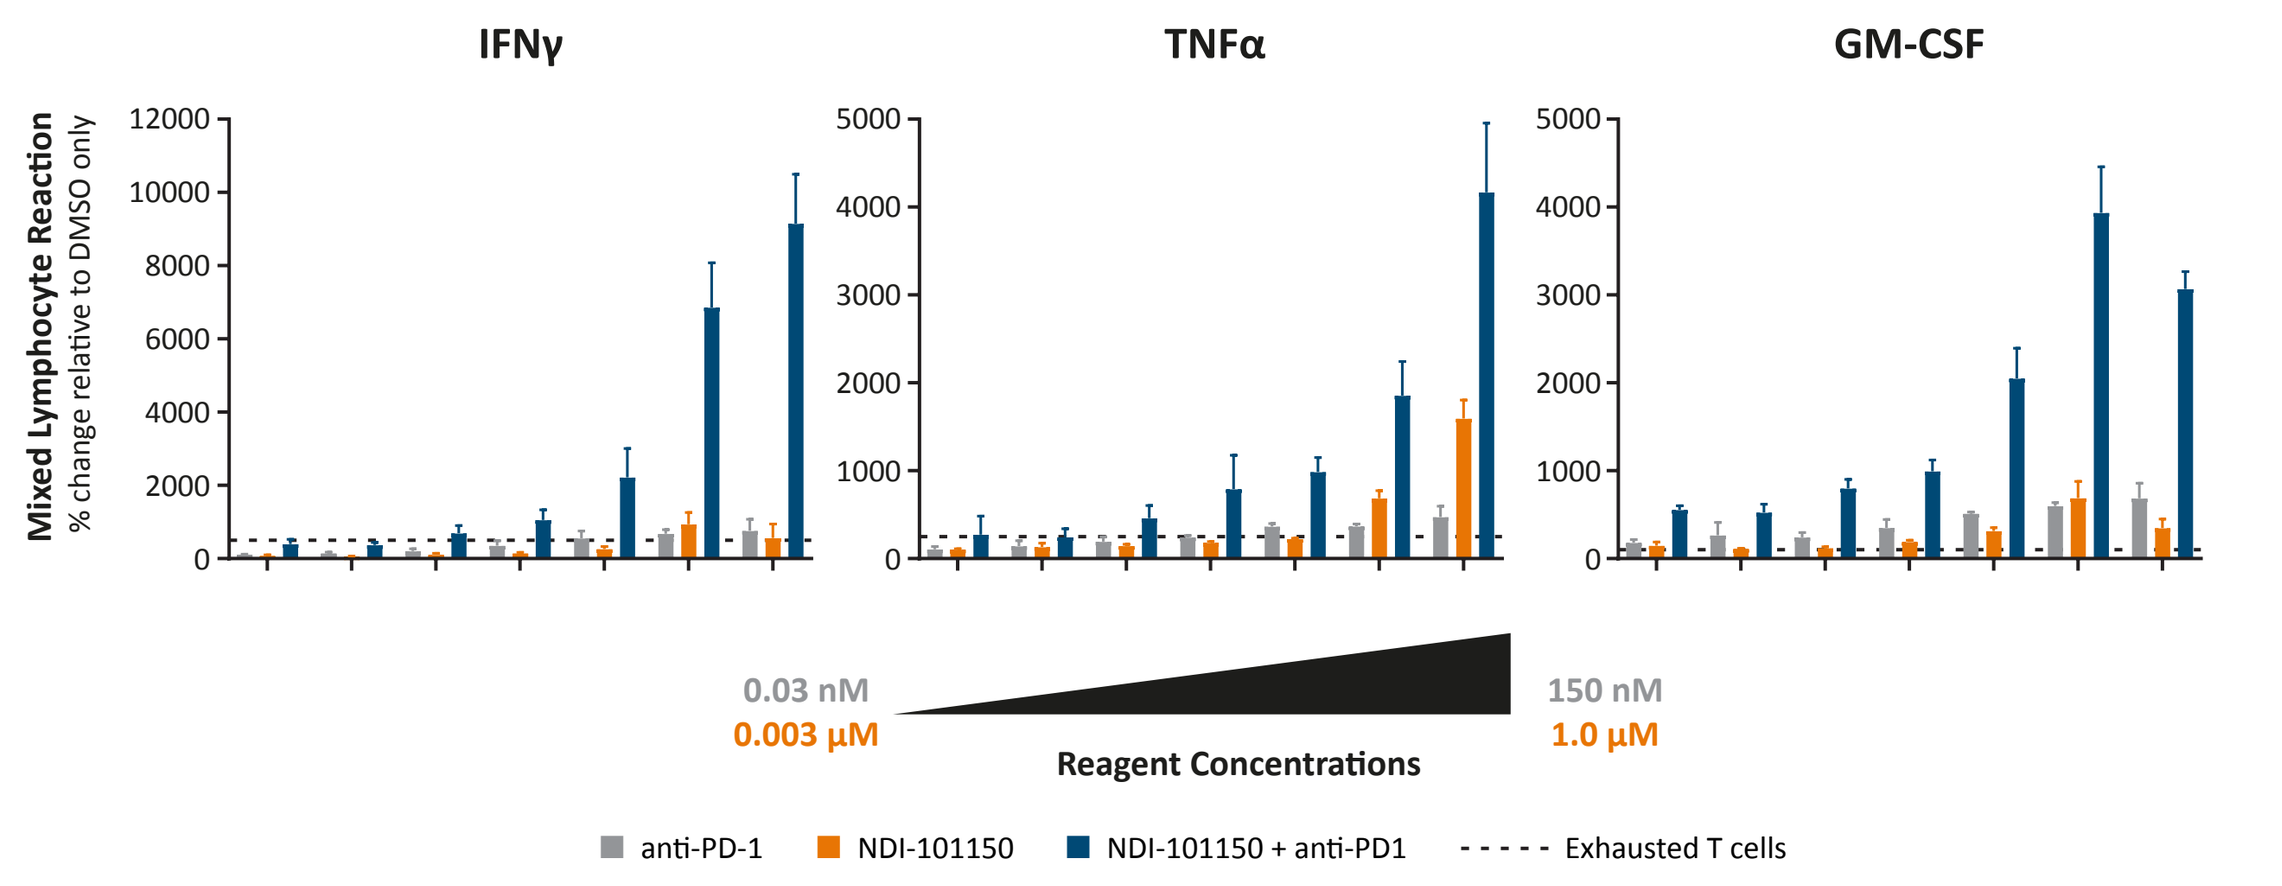
IFNγ
TNFα
GM-CSF
Mixed Lymphocyte Reaction
% change relative to DMSO only
0.03 nM
0.003 µM
Reagent Concentrations
150 nM
1.0 µM
anti-PD-1
NDI-101150
NDI-101150 + anti-PD1
Exhausted T cells

Figure 6. HPK1 Inhibition Overcomes Multiple Immunosuppressive Mechanisms

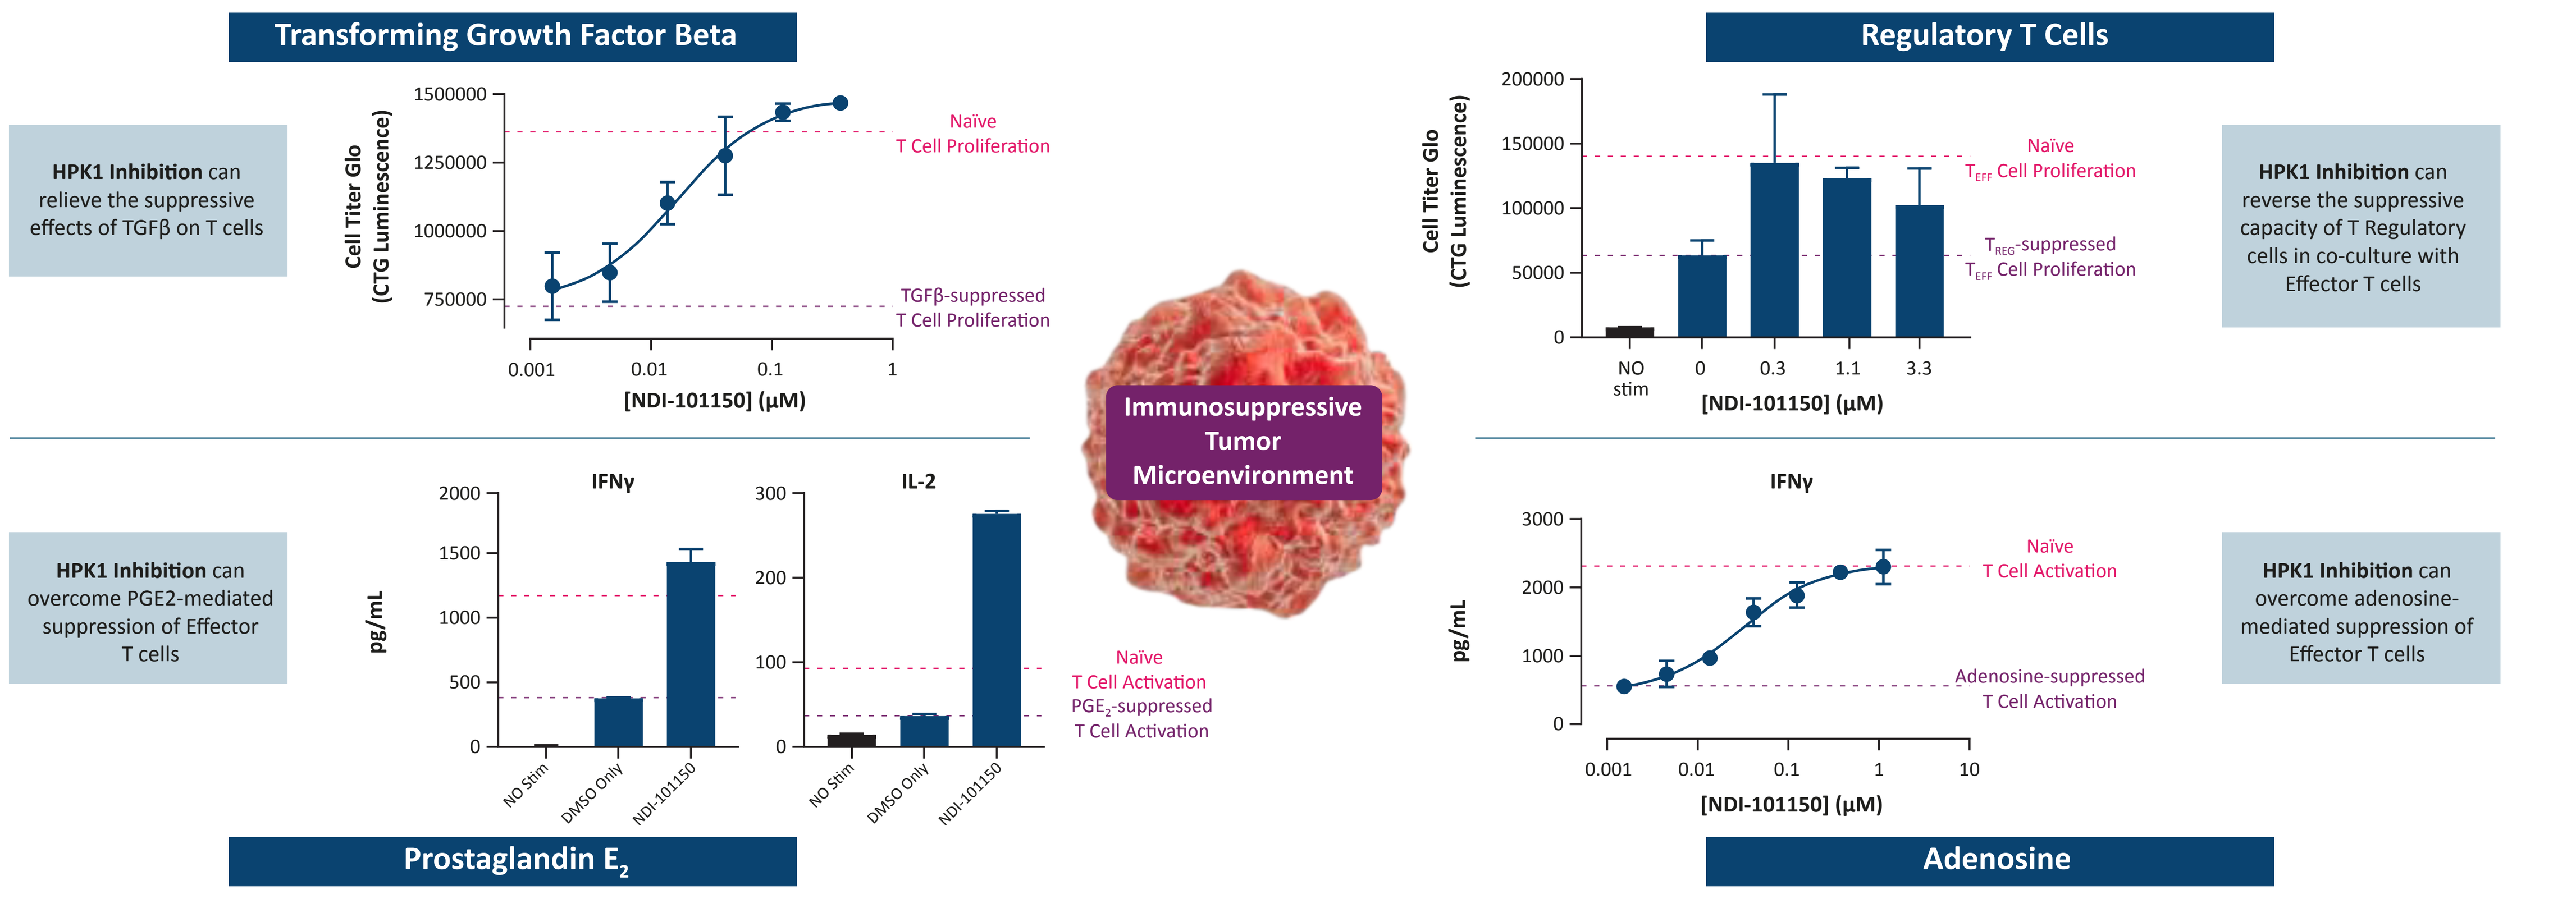
Transforming Growth Factor Beta
HPK1 Inhibition can relieve the suppressive effects of TGFβ on T cells
Cell Titer Glo (CTG Luminescence)
Naïve T Cell Proliferation
TGFβ-suppressed T Cell Proliferation
[NDI-101150] (µM)
Regulatory T Cells
Naïve T_{EFF} Cell Proliferation
T_{REG}-suppressed T_{EFF} Cell Proliferation
HPK1 Inhibition can reverse the suppressive capacity of T Regulatory cells in co-culture with Effector T cells
Immunosuppressive Tumor Microenvironment
HPK1 Inhibition can overcome PGE2-mediated suppression of Effector T cells
IFNγ
IL-2
pg/mL
NO Stim
DMSO Only
NDI-101150
Naïve T Cell Activation
PGE_2-suppressed T Cell Activation
Prostaglandin E_2
Naïve T Cell Activation
Adenosine-suppressed T Cell Activation
HPK1 Inhibition can overcome adenosine-mediated suppression of Effector T cells
Adenosine

Figure 7. NDI-101150 Treatment Enhances the Activity of Human B Cells

B cells were purified from human donors, pre-treated with various concentrations of NDI-101150 or DMSO for 1 hour, and then stimulated. Expression of CD69 on the surface of B cells was determined by FACS at 2 hours post-stimulation (purple bars), IgG production from B cells was quantified by ELISA at 4 days post-stimulation (blue bars), and proliferation of B cells was measured using CellTiter Glo® at 5 days post-stimulation (orange bars).

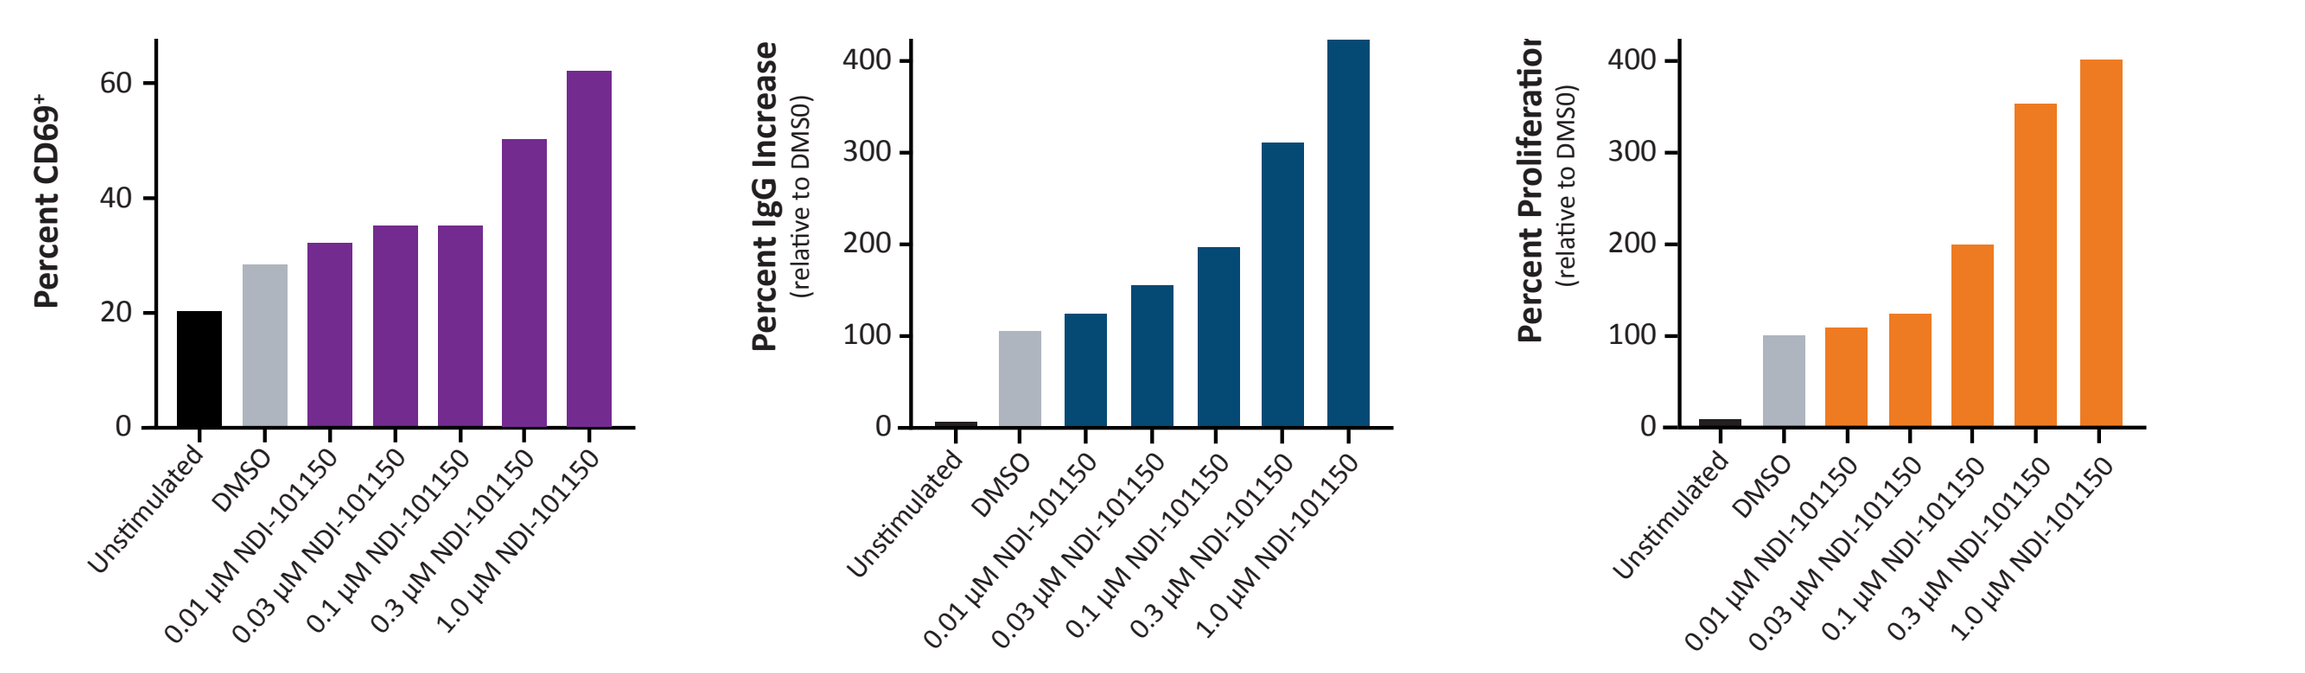
Percent CD69+
Percent IgG Increase (relative to DMSO)
Percent Proliferation (relative to DMSO)
Unstimulated
DMSO
0.01 µM NDI-101150
0.03 µM NDI-101150
0.1 µM NDI-101150
0.3 µM NDI-101150
1.0 µM NDI-101150

Figure 8. NDI-101150 Enhances Cytokine Secretion, Co-Stimulatory Receptor Expression, and Antigen-Presentation Capacity of DCs

Cytokine production via ELISA (orange bars) and costimulatory receptor expression via FACS (light blue bars) were measured from bone marrow-derived dendritic cells (BMDCs) matured in the presence of various concentrations of NDI-1011502 or DMSO. BMDCs pulsed with OVA peptide and cultured in the presence of NDI-101150 were assayed for their ability to activate purified OTI CD8+ T cells (blue bars).

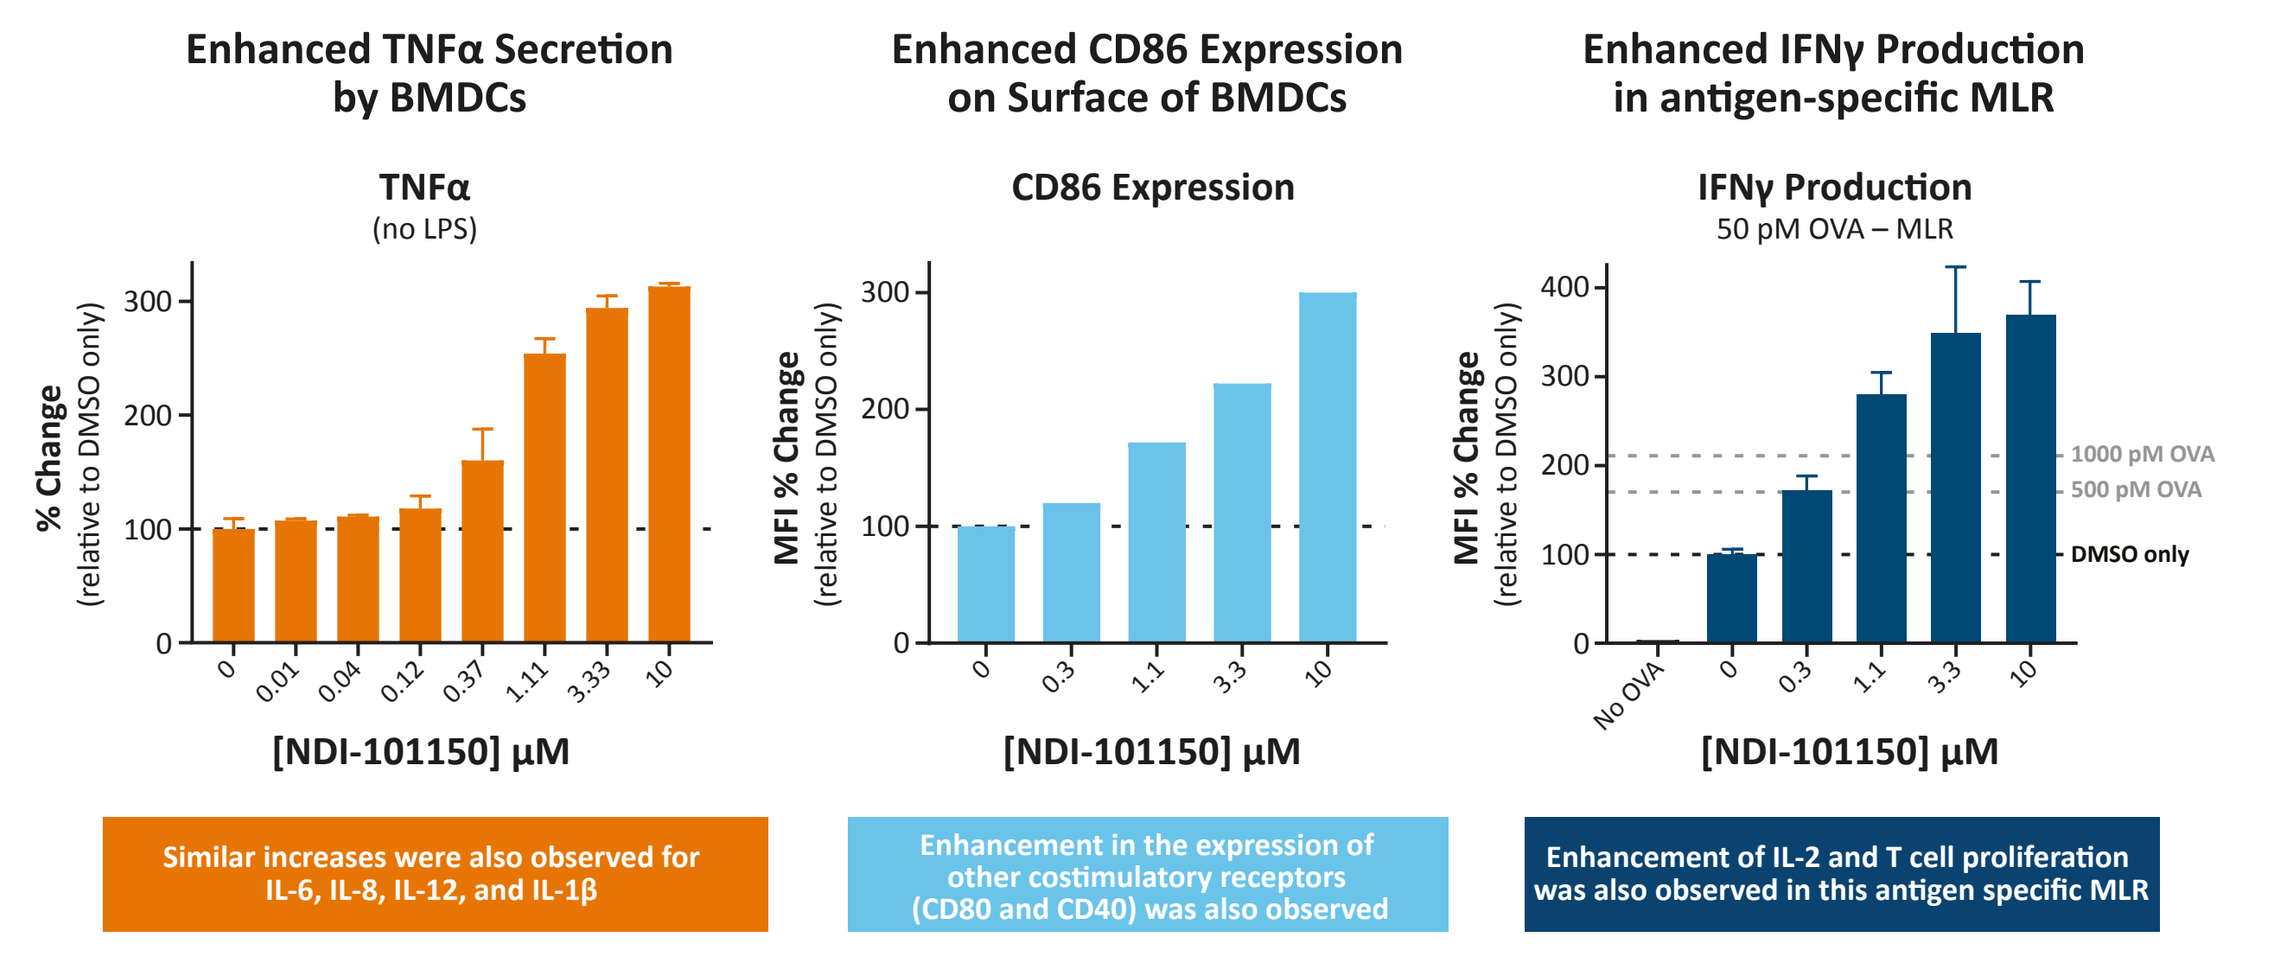
Enhanced TNFα Secretion by BMDCs
TNFα
(no LPS)
% Change (relative to DMSO only)
[NDI-101150] µM
Similar increases were also observed for IL-6, IL-8, IL-12, and IL-1β
Enhanced CD86 Expression on Surface of BMDCs
CD86 Expression
MFI % Change (relative to DMSO only)
Enhancement in the expression of other costimulatory receptors (CD80 and CD40) was also observed
Enhanced IFNγ Production in antigen-specific MLR
IFNγ Production
50 pM OVA – MLR
1000 pM OVA
500 pM OVA
DMSO only
No OVA
Enhancement of IL-2 and T cell proliferation was also observed in this antigen specific MLR

Figure 9. NDI-101150 Treatment Induces Robust Tumor Growth Inhibition and a Durable Immune Memory Response in EMT-6 Syngeneic Mouse Model

NDI-101150 treatment resulted in multiple complete responses (CRs, no measurable tumor) in the EMT-6 syngeneic model. Compound-dosing was discontinued in these animals and after the 5-day washout period, animals were re-challenged with EMT6 cells in the opposite flank and tumor growth was monitored for an additional 28 days (no further treatment was administered).

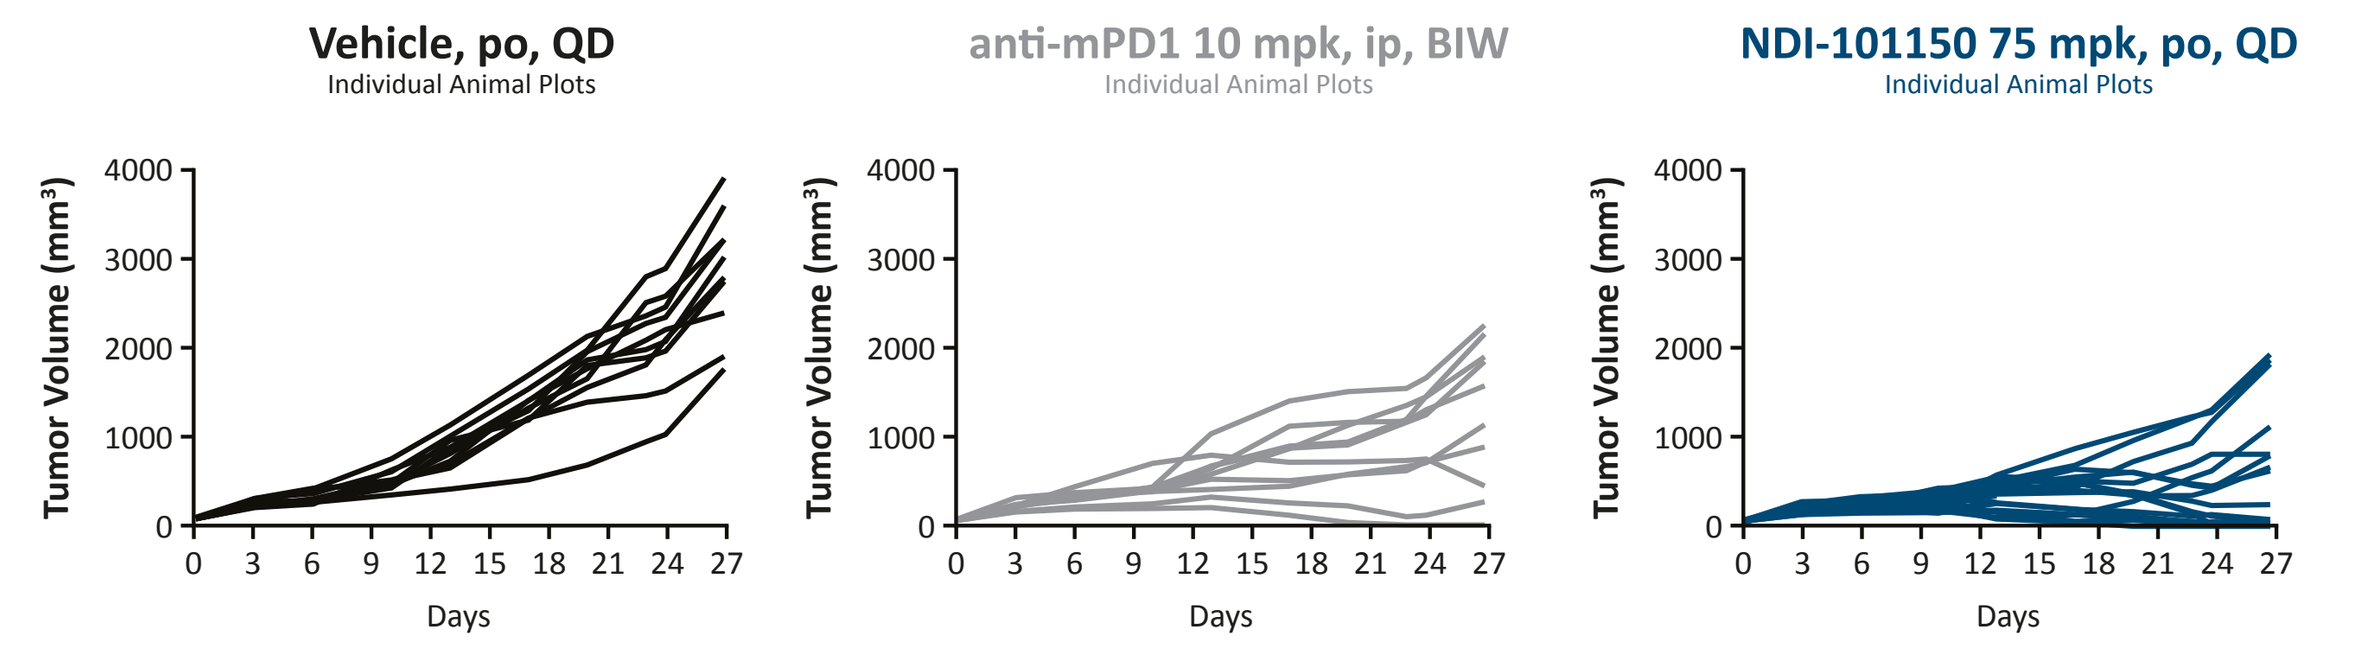
Vehicle, po, QD
Individual Animal Plots
anti-mPD1 10 mpk, ip, BIW
Individual Animal Plots
NDI-101150 75 mpk, po, QD
Individual Animal Plots
Tumor Volume (mm³)
Days

Treatment Group	P-Value (T-test)	CRs
Vehicle, po, QD		
Anti-mPD1 10 mpk, ip, BIW	0.0002	1
NDI-101150 75 mpk, po, QD	< 0.0001	6

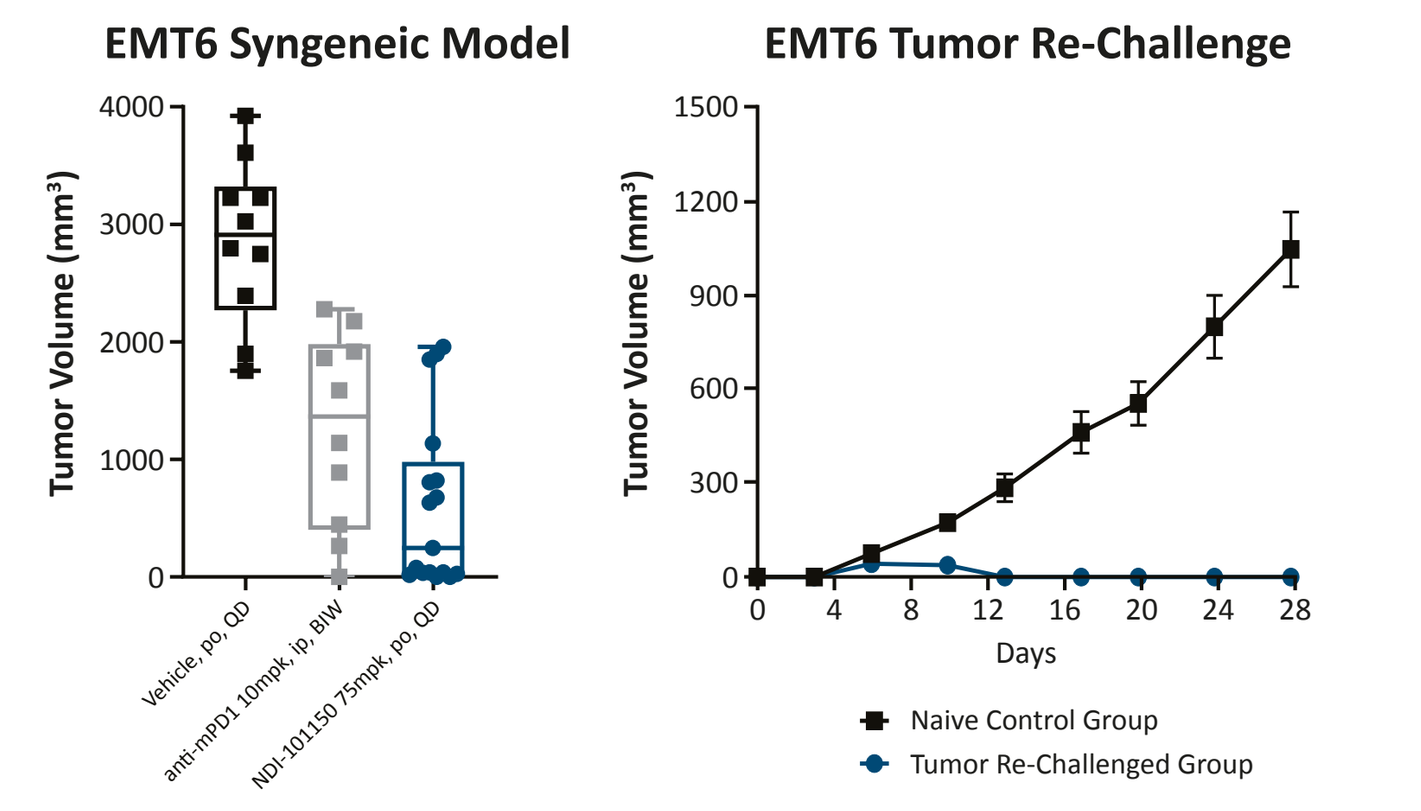
EMT6 Syngeneic Model
Tumor Volume (mm³)
Vehicle, po, QD
anti-mPD1 10mpk, ip, BIW
NDI-101150 75mpk, po, QD
EMT6 Tumor Re-Challenge
Days
Naive Control Group
Tumor Re-Challenged Group

Figure 10. NDI-101150-Treated Murine EMT-6 Tumors are Infiltrated with Cytotoxic CD8+ T cells, CD19+ B cells, and Dendritic Cells

Mice were injected SC with EMT-6 tumor cells into the right flank and randomized once a mean tumor volume of 100 mm^3 was reached. Tumor-infiltrating lymphocytes were identified and characterized by FACS and multiplex immunofluorescence.

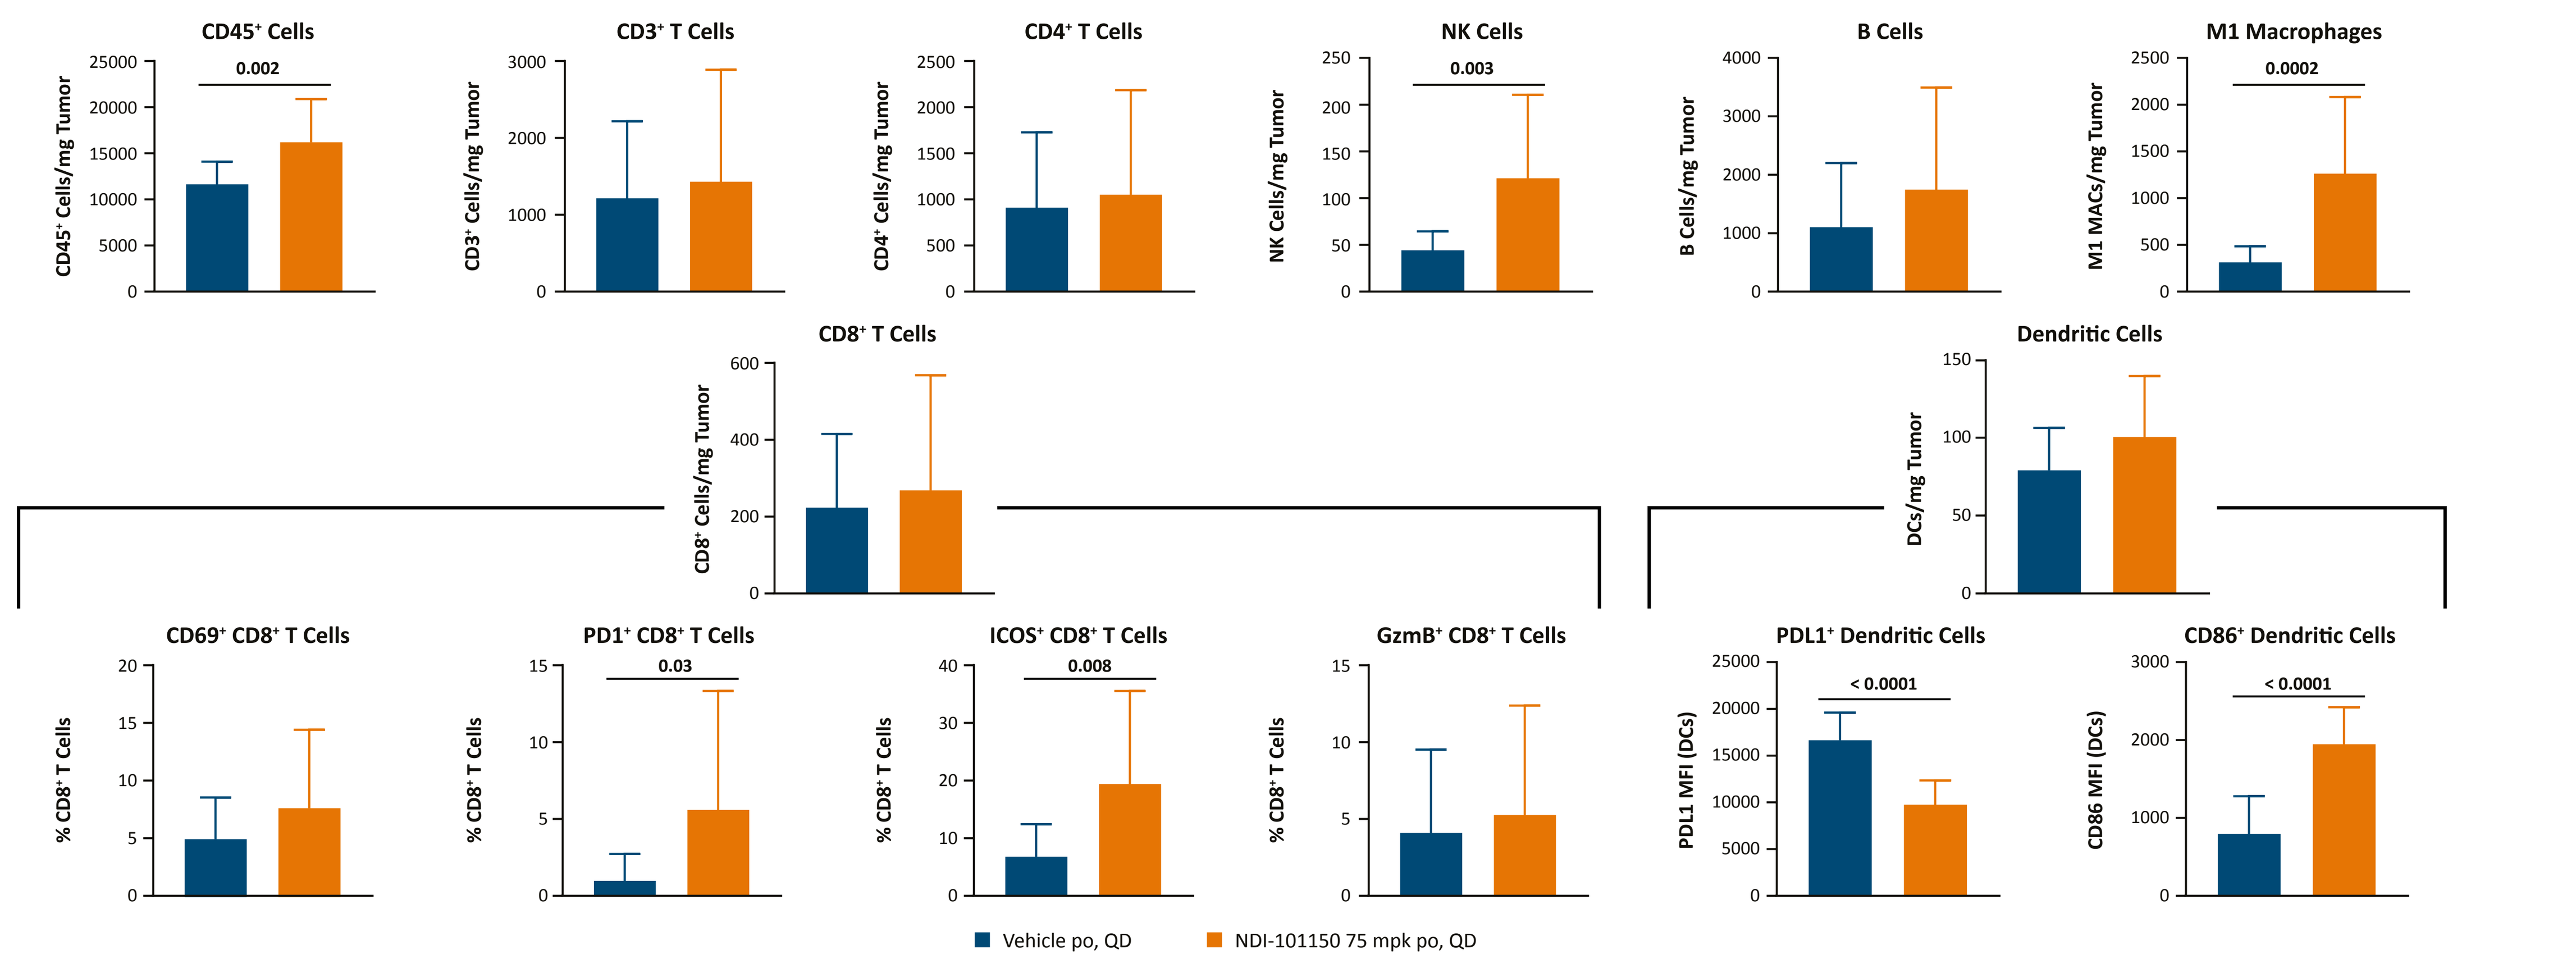
CD45+ Cells
0.002
CD3+ T Cells
CD4+ T Cells
NK Cells
0.003
B Cells
M1 Macrophages
0.0002
CD8+ T Cells
Dendritic Cells
CD69+ CD8+ T Cells
PD1+ CD8+ T Cells
0.03
ICOS+ CD8+ T Cells
0.008
GzmB+ CD8+ T Cells
PDL1+ Dendritic Cells
< 0.0001
CD86+ Dendritic Cells
< 0.0001
Vehicle po, QD
NDI-101150 75 mpk po, QD

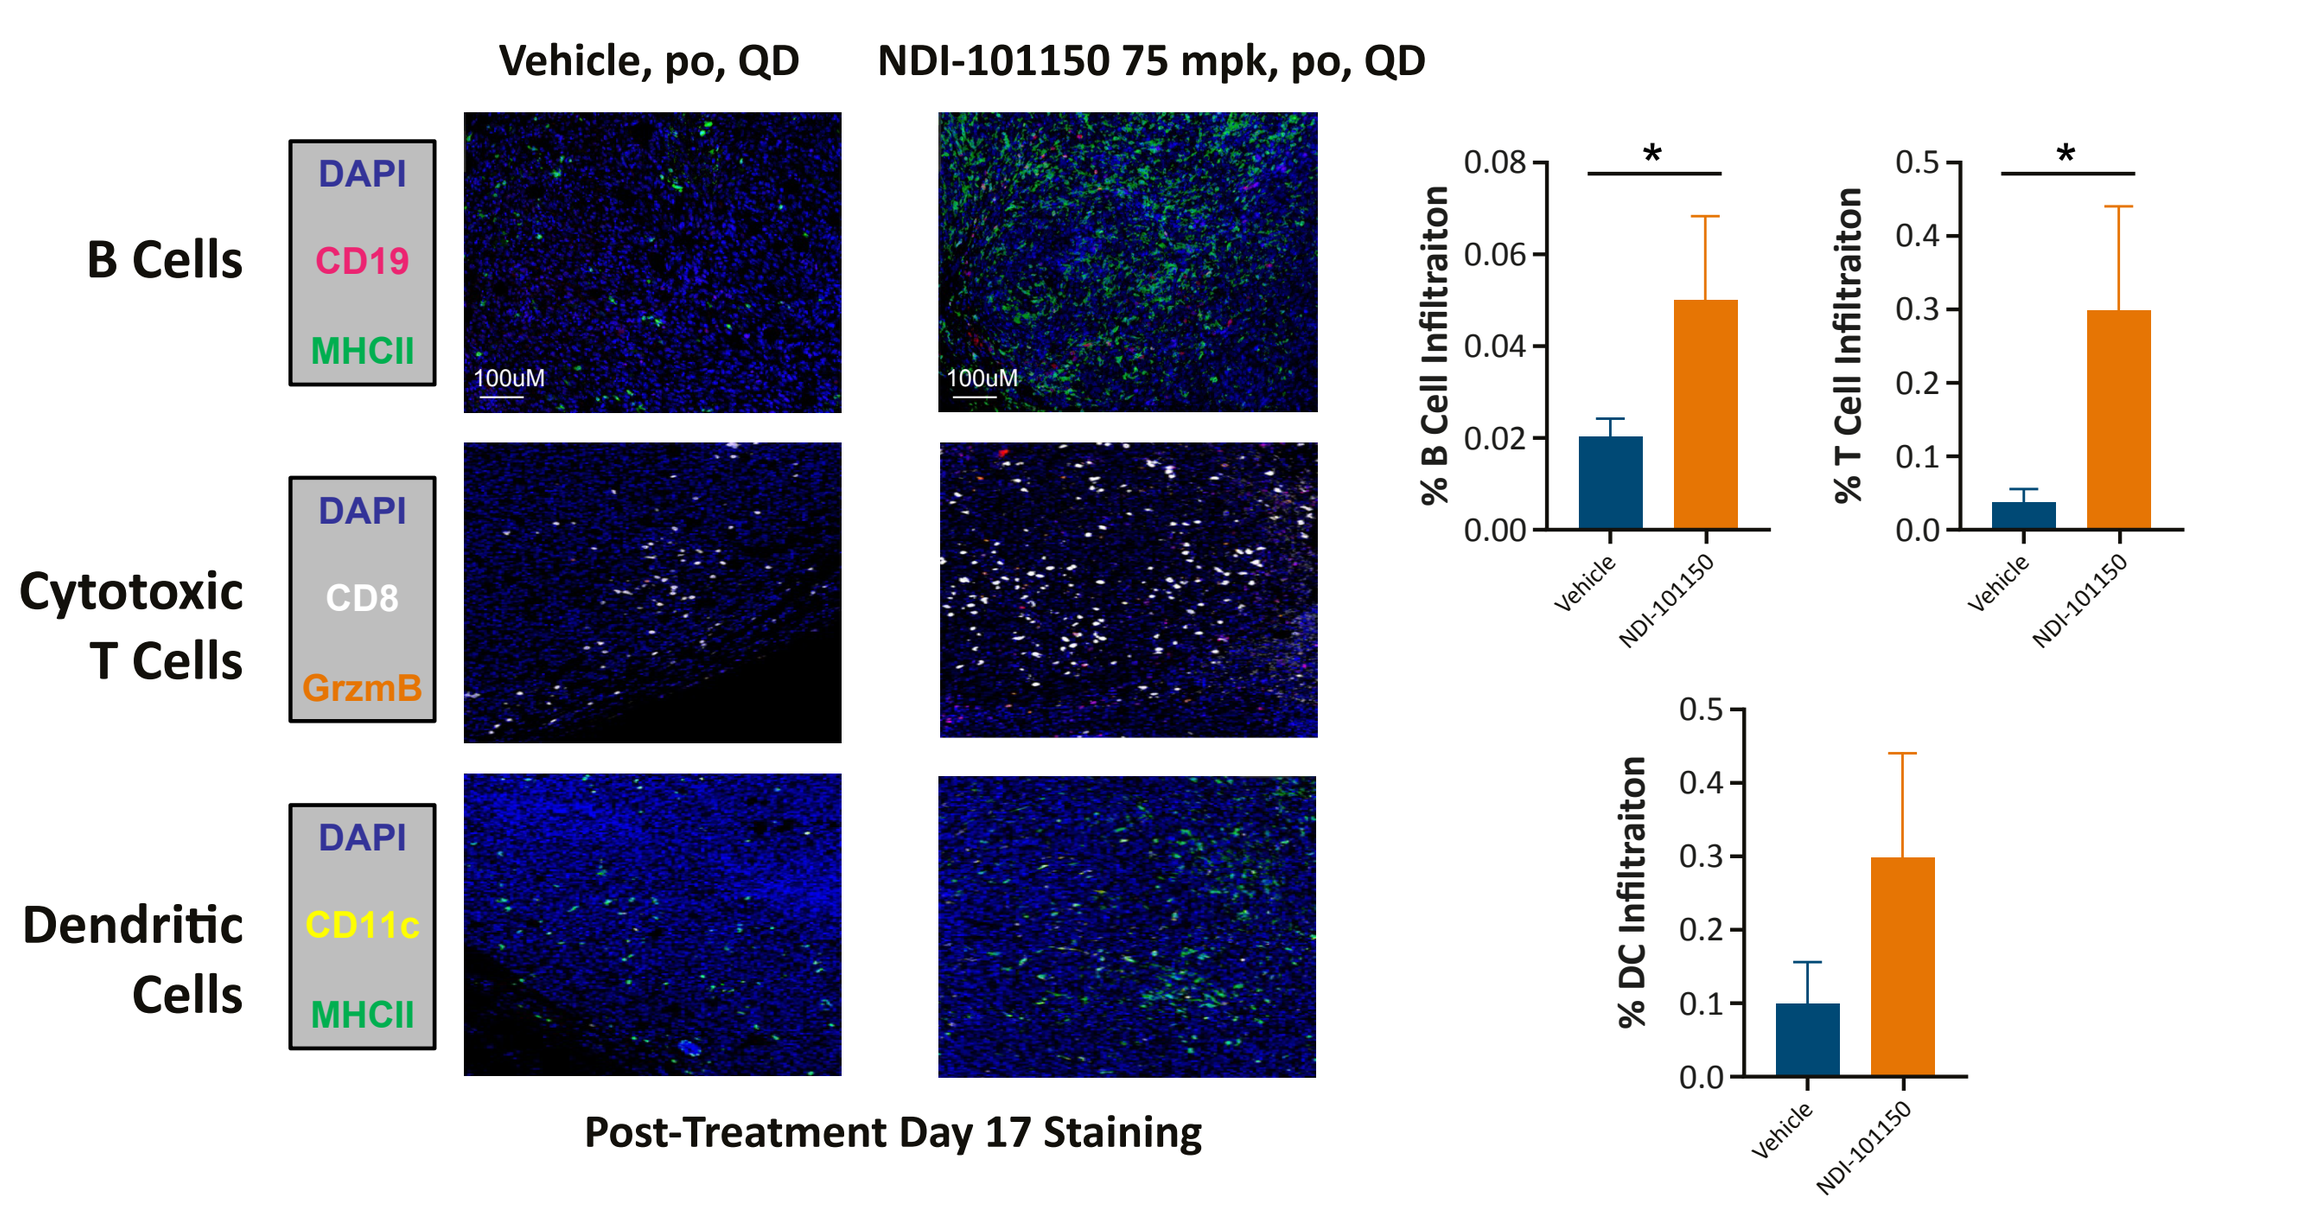
Vehicle, po, QD
NDI-101150 75 mpk, po, QD
B Cells
DAPI
CD19
MHCII
Cytotoxic T Cells
DAPI
CD8
GrzmB
Dendritic Cells
DAPI
CD11c
MHCII
% B Cell Infiltraiton
% T Cell Infiltraiton
% DC Infiltraiton
Vehicle
NDI-101150

Post-Treatment Day 17 Staining

CONCLUSIONS

- The data we present support the use of NDI-101150 in creating a powerful multifaceted antitumor immune response alone or in combination with immune checkpoint inhibitor therapies
- NDI-101150 is currently being investigated in a first-in-human multi-center open-label phase 1/2 trial (NCT05128487) as monotherapy or in combination with pembrolizumab in patients with advanced solid tumors.

[a]Presenting author: david.ciccone@nimbustx.com

Acknowledgements
Editorial assistance was provided by Melody Watson, Bioscript Group, Macclesfield, UK, and supported by Nimbus Therapeutics (Nimbus Discovery Inc. on behalf of Nimbus Saturn Inc.)

Disclosures
DC, F-S K, SB, SD, SC, NK, GY, EAG, DL, BS, FGB, and **CL** are employees of Nimbus Therapeutics (Nimbus Discovery Inc. on behalf of Nimbus Saturn Inc.)

References
1. Alzabin S., et al. *Cancer Immunology and Immunotherapy* 2010, v59: 419-429; 2. Hernandez S., et al. *Cell Reports* 2018. v25: 80-94. 3. Ciccone et al; AACR-NCI-EORTC 2023 meeting poster C065. 4. Sommerhalder et al. SITC 2023

Presented at the Society for Immunotherapy of Cancer 38th Annual Conference, November 1–5, 2023