Acetyl-CoA Carboxylase Inhibition by ND-630 Inhibits Fatty Acid Synthesis, Stimulates Fatty Acid Oxidation, Reduces Body Weight, Improves Insulin Sensitivity, and Modulates Dyslipidemia in Rats

Geraldine Harriman, Jeremy Greenwood, Sathesh Bhat, Rosana Kapeller, and H. James Harwood Jr.

Nimbus Discovery, Cambridge, MA and Schrodinger, New York, NY (USA)

ABSTRACT

Simultaneous inhibition of the acetyl-CoA carboxylase isozymes, ACC1 & ACC2, results in concomitant inhibition of fatty acid synthesis (FASyn) and stimulation of fatty acid oxidation (FAOxn) and may favorably affect obesity, diabetes, and fatty liver disease. Our efforts to discover ACC inhibitors have focused on interaction with the subunit dimerization site on the biotin carboxylase (BC) domain of the enzyme to which the phosphopeptide of phosphoACC binds to prevent dimerization and to which the fungal metabolite Soraphen A interacts. Using state-of-the-art structure-based drug design and crystal structures of human ACC2 BC domain, we identified a unique series of allosteric inhibitors that bind to the Soraphen binding site, inhibit enzymatic activity, exhibit functional activity in cultured cells, and exhibit favorable drug-like properties and *in vivo* efficacy. For example, NDI-630 inhibits human ACC1&2 (IC_{50} 2.0 nM), inhibits HepG2 cell FASyn (EC_{50} 66 nM), stimulates C2C12 cell FAOxn (2-fold 200 nM), inhibits rat liver FASyn (ED_{50} 0.14 mg/kg), and stimulates rat whole body FAOxn (MED 3 mg/kg). When administered orally 1x/day for 14 days to high fat-fed diet-induced obese (DIO) rats at doses 3-30 mg/kg, NDI-630 reduced weight gain without affecting food intake, reduced the hyperleptinemia and hyperinsulinemia produced by the high fat diet without altering plasma glucose, improved insulin sensitivity (oral glucose tolerance testing), and reduced hepatic triglycerides to chow-fed control levels. When administered as above for 28 days to high sucrose-fed DIO rats, in addition to the above effects, NDI-630 also reduced plasma triglycerides and free fatty acids to chow-fed control levels and markedly reduced plasma cholesterol. These observations suggest that our allosteric ACC inhibitors may favorably affect obesity, diabetes, and fatty liver disease.

FIGURE 1: Acetyl CoA Carboxylase (ACC): Master Regulator of Fatty Acid Synthesis & Oxidation

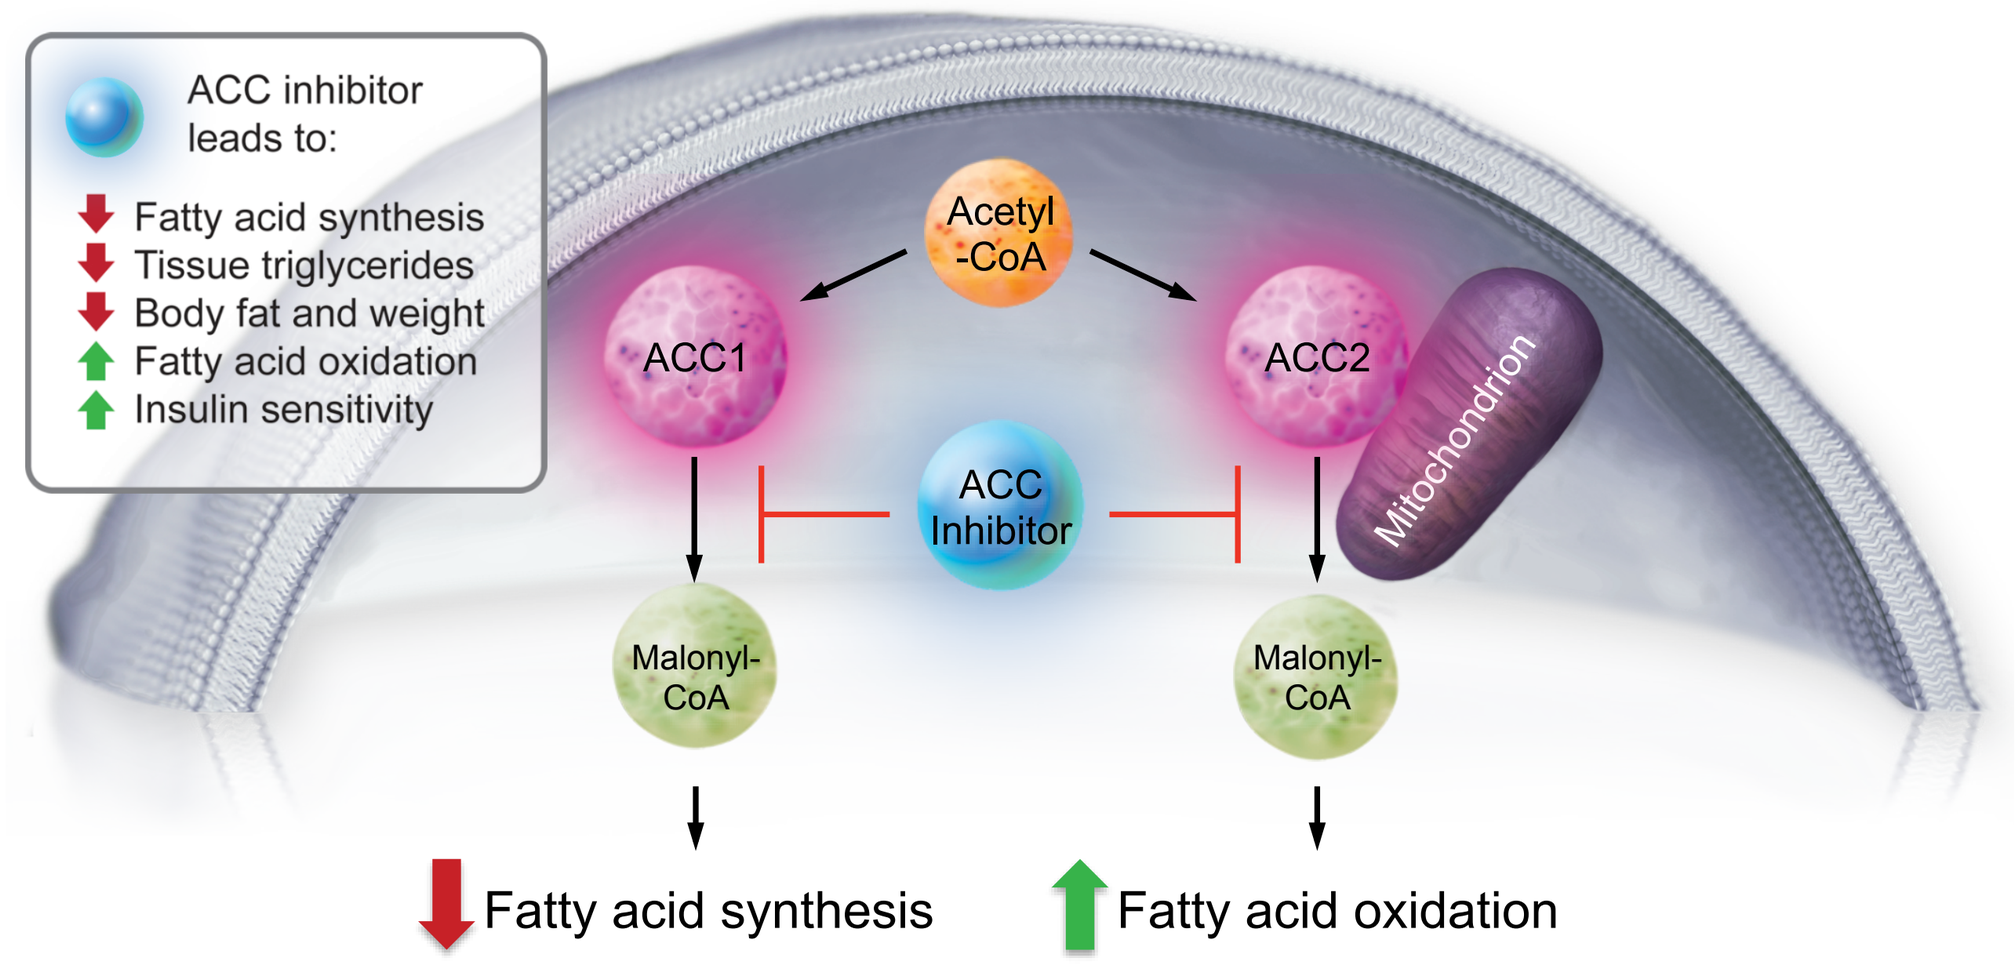
ACC inhibitor leads to:
Fatty acid synthesis
Tissue triglycerides
Body fat and weight
Fatty acid oxidation
Insulin sensitivity
Acetyl -CoA
ACC1
ACC2
ACC Inhibitor
Mitochondrion
Malonyl-CoA
Malonyl-CoA
Fatty acid synthesis
Fatty acid oxidation

- Beneficial effects on lipids, blood glucose, weight, potentially diabetes and CV risk.
- Nimbus: **First** small molecule **allosteric** inhibitor successfully targeting BC domain.

FIGURE 2: Nimbus' Unique Approach Identified Potent ACC BC Domain Inhibitors Targeting Soraphen Binding Site

- Nimbus took an orthogonal approach and designed inhibitors that bound to the Soraphen binding site in the BC domain of the ACC complex.

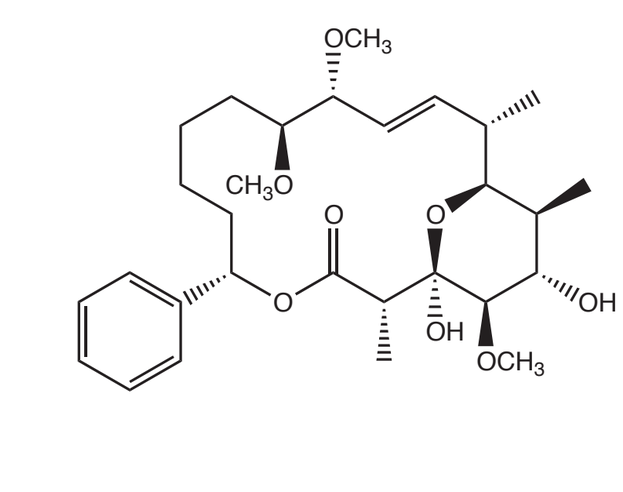
OCH3
CH3O
OH
OH
OCH3

Soraphen A

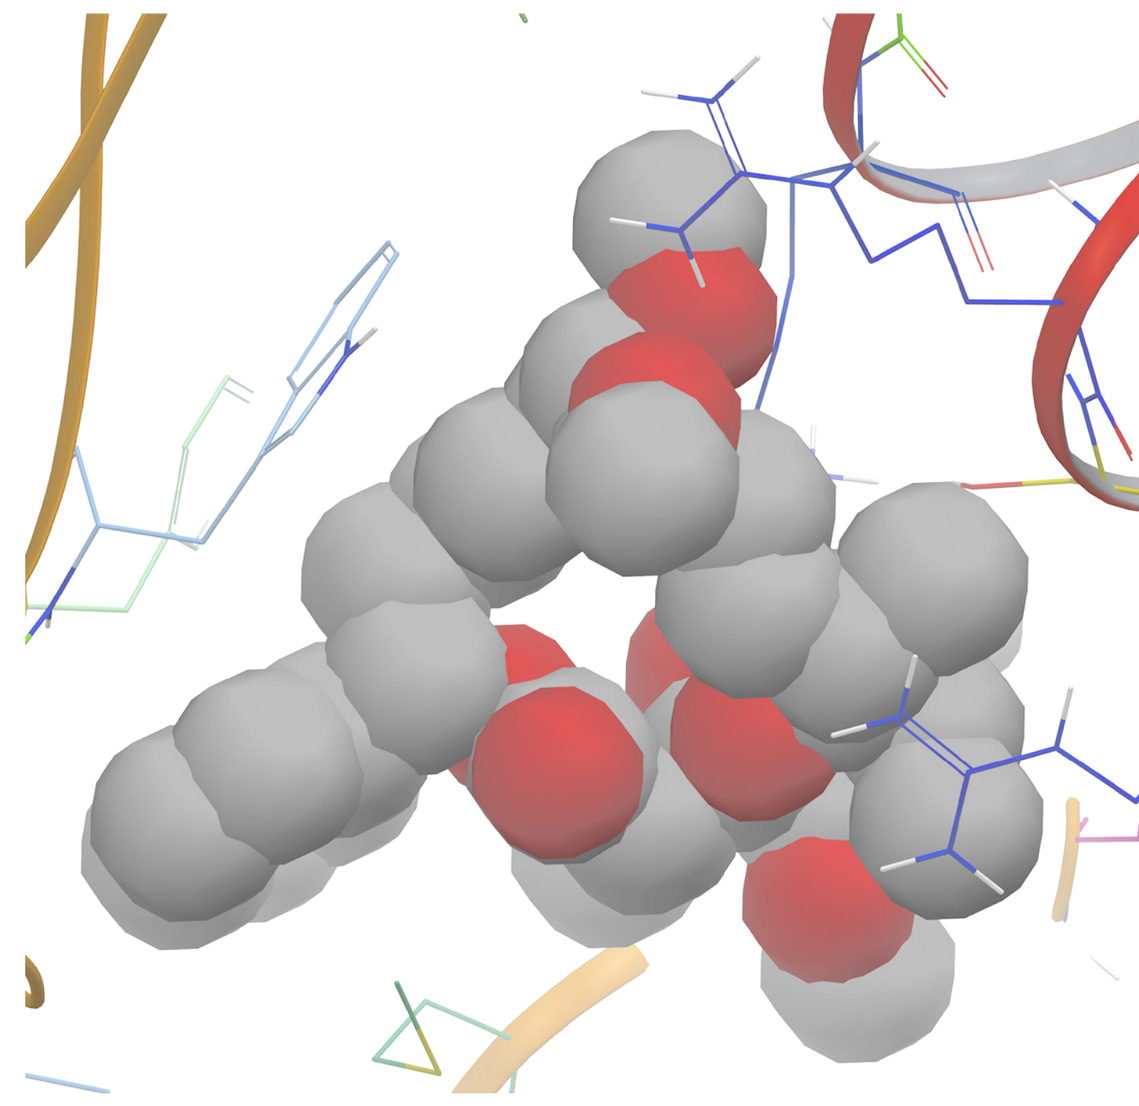

FIGURE 3: Nimbus Approach has Delivered ACC Inhibitors with Demonstrated *in Vivo* Efficacy in 16 Months

- A structure-based virtual screen of 1.3M lead-like molecules was performed utilizing Schrodinger's computational tools including WaterMap and Glide. This led to the identification of a high quality family of hit molecules with micromolar inhibitory effects against the enzyme. Co-crystal structures of members of this series in the human BC domain confirmed the model and our approach.

Ideas	1.3MM	5,000	10,000
Total synthesized compound		175	225
Potency ACC1, ACC2	**8,000 nM**	**<10 nM**	**<1 nM**

Milestone Achieved

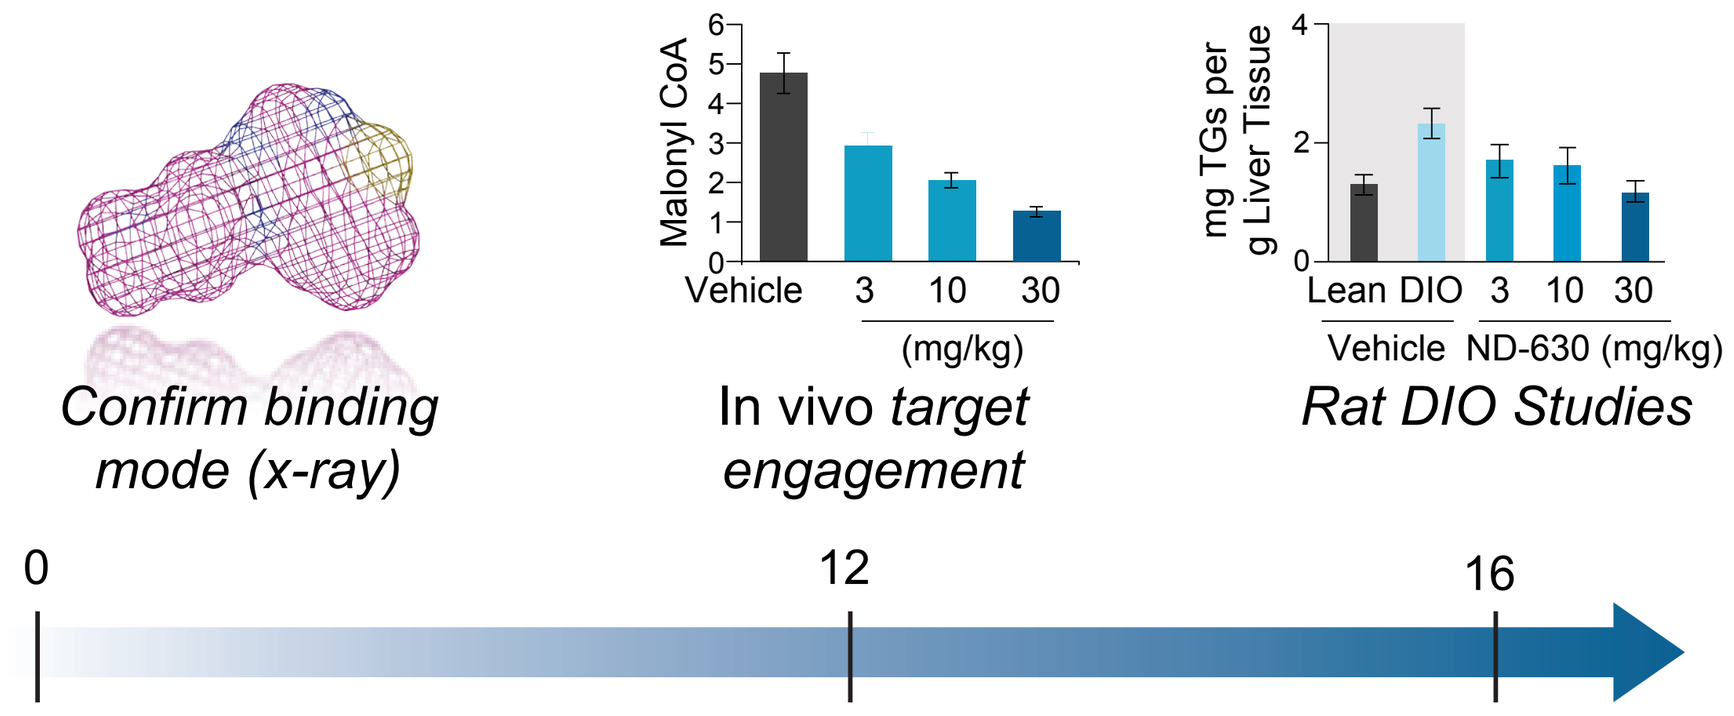
Malonyl CoA
Vehicle 3 10 30
(mg/kg)
mg TGs per g Liver Tissue
Lean DIO 3 10 30
Vehicle ND-630 (mg/kg)
Confirm binding mode (x-ray)
In vivo target engagement
Rat DIO Studies
0
12
16
Months

- In an iterative design fashion over the next 16 month period, the potency of this family of hits were improved 1000x utilizing the computational model focusing on the replacement or stabilization of high energy hydration sites within the Soraphen binding site.
- Simultaneous to the potency improvements, drug-like properties were optimized to deliver Development Candidate quality molecules that have demonstrated activity in chronic models of metabolic syndrome and diabetes.

FIGURE 4: ND-630 Displays Excellent Drug-like Properties

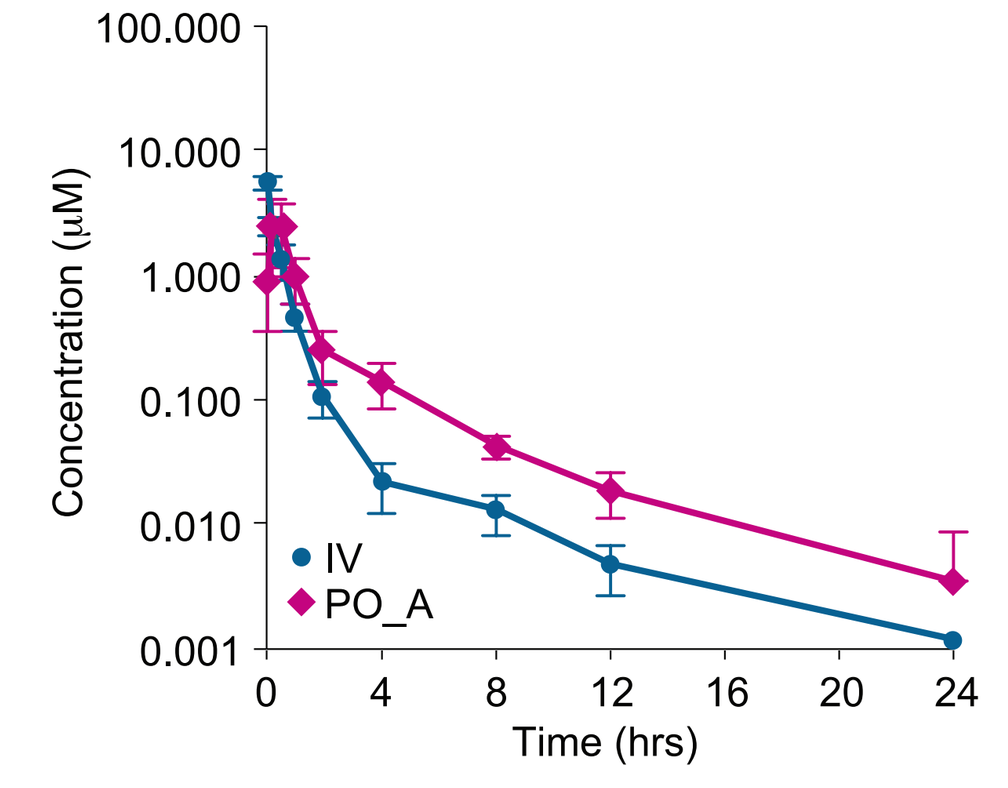
Plasma PK Profile
SD Rats IV (3 mg/kg, n=3, solution dose)
PO_A (10 mg/kg, n=3, suspension dose)
Concentration (µM)
IV
PO_A
Time (hrs)

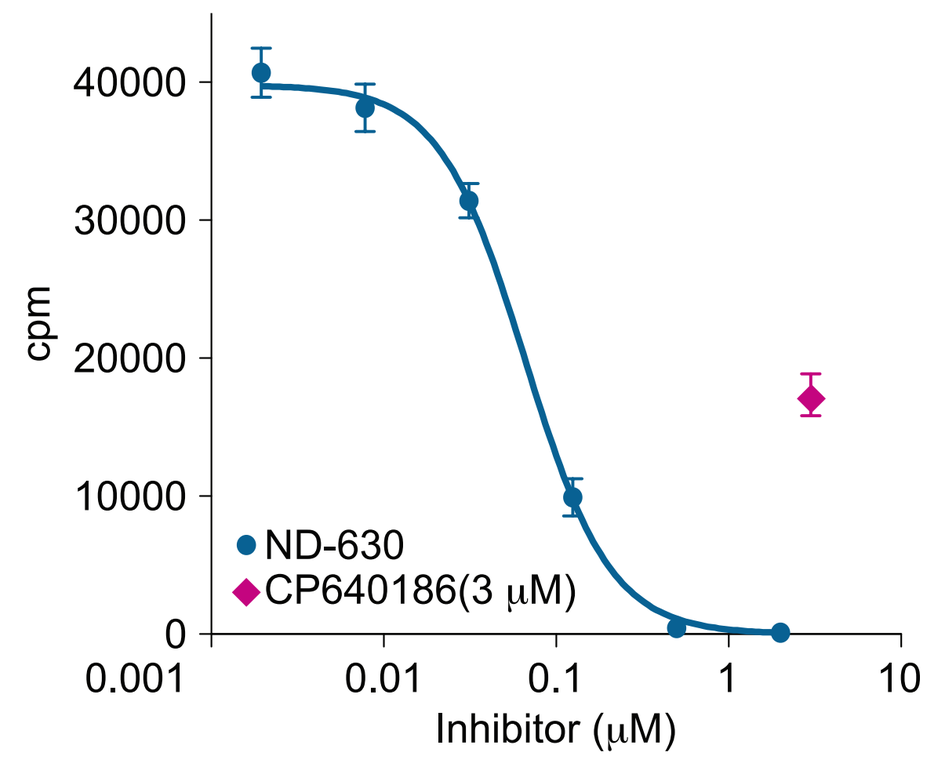
Hep-G2 cell FAsyn Inhibition
[14C]acetate incorporation into FA)
cpm
ND-630
CP640186(3 µM)
Inhibitor (µM)

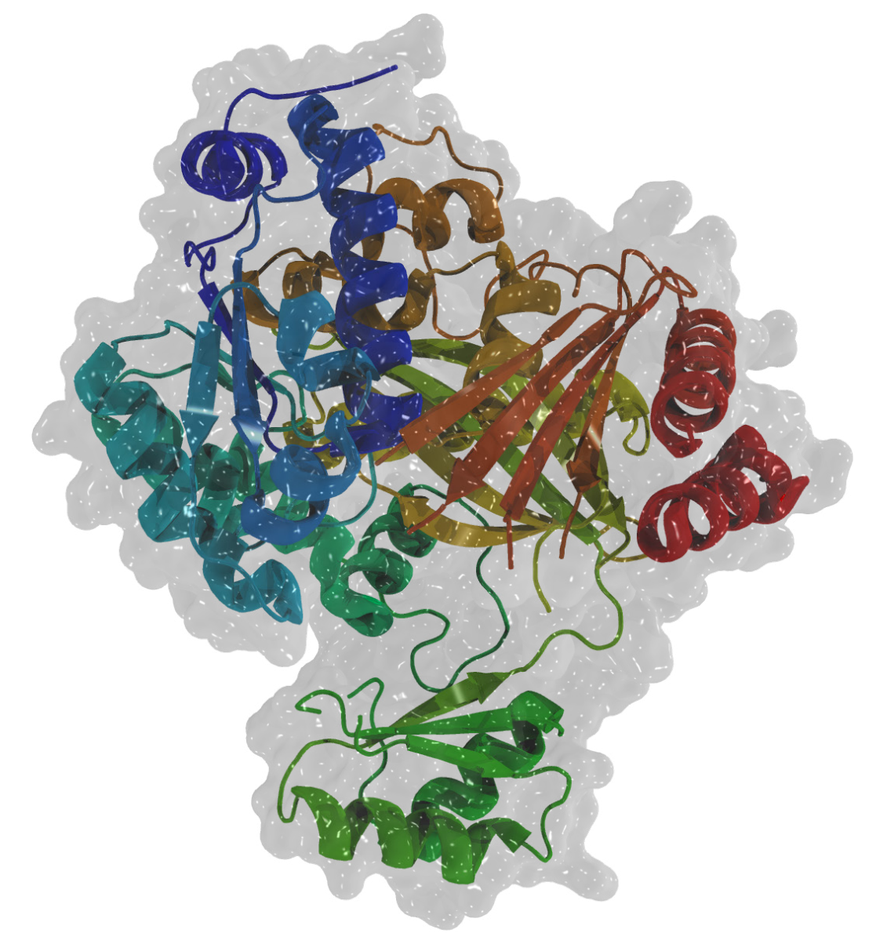
Confirmatory Co-crystal Structures

TARGET POTENCY
- ACC1 IC_{50} = 2 nM
- ACC2 IC_{50} = 2 nM
- HepG2 EC_{50} = 66 nM
- C2C12 FAOxn stim. 2x @ 200 nM

ADMET
- Low multispecies intrinsic clearance (human, mouse, rat, dog, monkey)
- High solubility (>300 µM)
- P450 inhib >50 µM, hERG >30 µM
- Protein binding (98%)
- DrugMatrixPanel 0/120 hits; 1000x window

RAT PK (10 mg/kg, PO)
- 37%F, Cmax 6 µM, Cl 33 mL/min/kg, AUC 3.4 µM.hr, T1/2 = 4.5h, Vss 2 L/kg
- Liver exposure: 81 µM
- Muscle exposure: 0.45 µM

PHARMACOLOGY
- Rat RQ MED = 3 mpk PO
- Rat FASyn ED_{50} = 0.14 mg/kg, PO
- Rat Malonyl-CoA ED_{50} = 0.15 mg/kg, PO

FIGURE 5: Proof of Mechanism:

ND-630 Acutely Inhibits ACC Prevents Malonyl CoA Production in the Liver (ED_{50}~1.5 mg/kg)

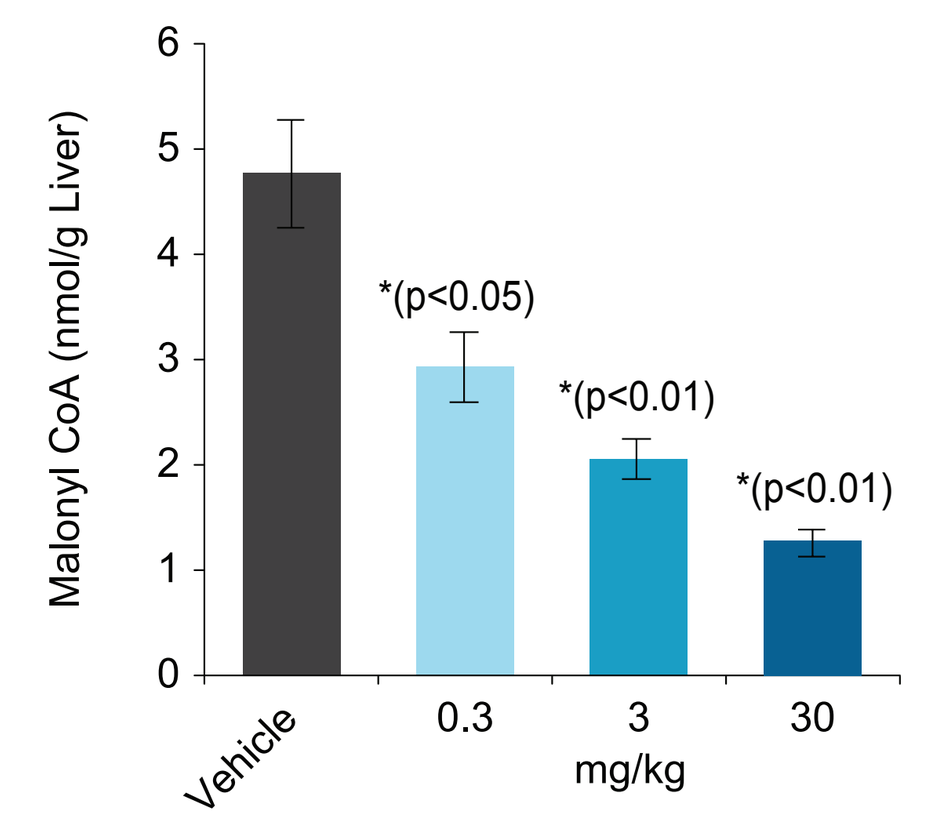
Malonyl CoA (nmol/g Liver)
*(p<0.05)
*(p<0.01)
*(p<0.01)
Vehicle
0.3
3
30
mg/kg

FIGURE 6: ND-630 *in vivo* Proof of Concept:

Target Engagement in the Liver and Muscle

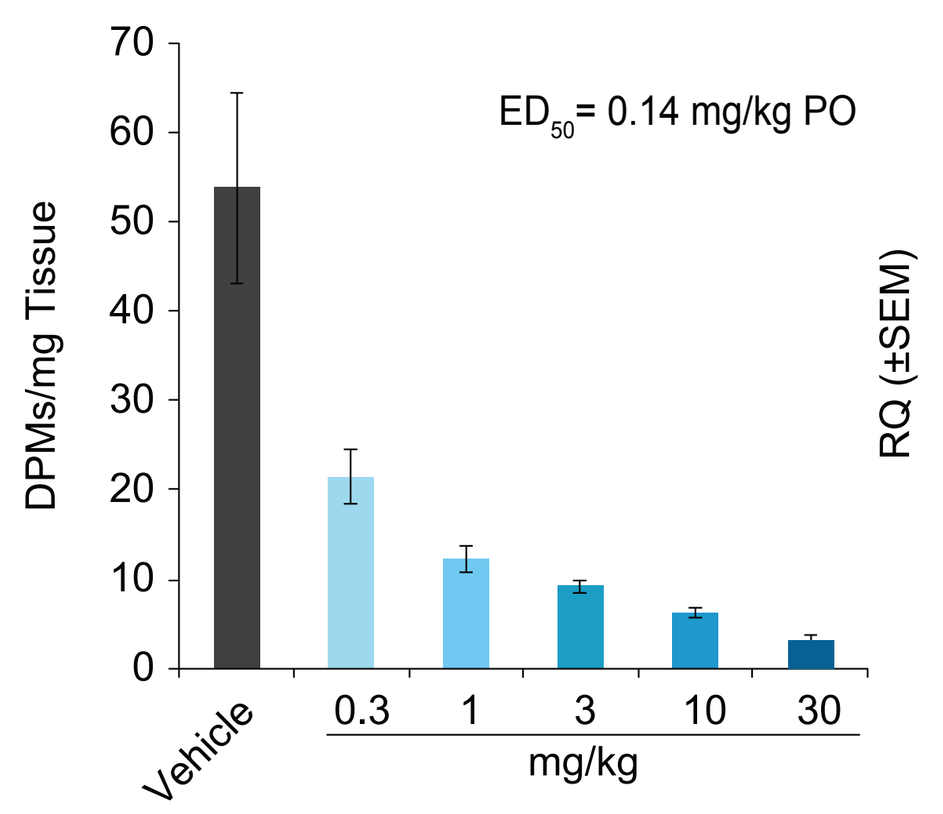
Target Engagement in Liver
(Fatty acid synthesis inhibition)
ED50= 0.14 mg/kg PO
DPMs/mg Tissue
Vehicle
0.3 1 3 10 30
mg/kg

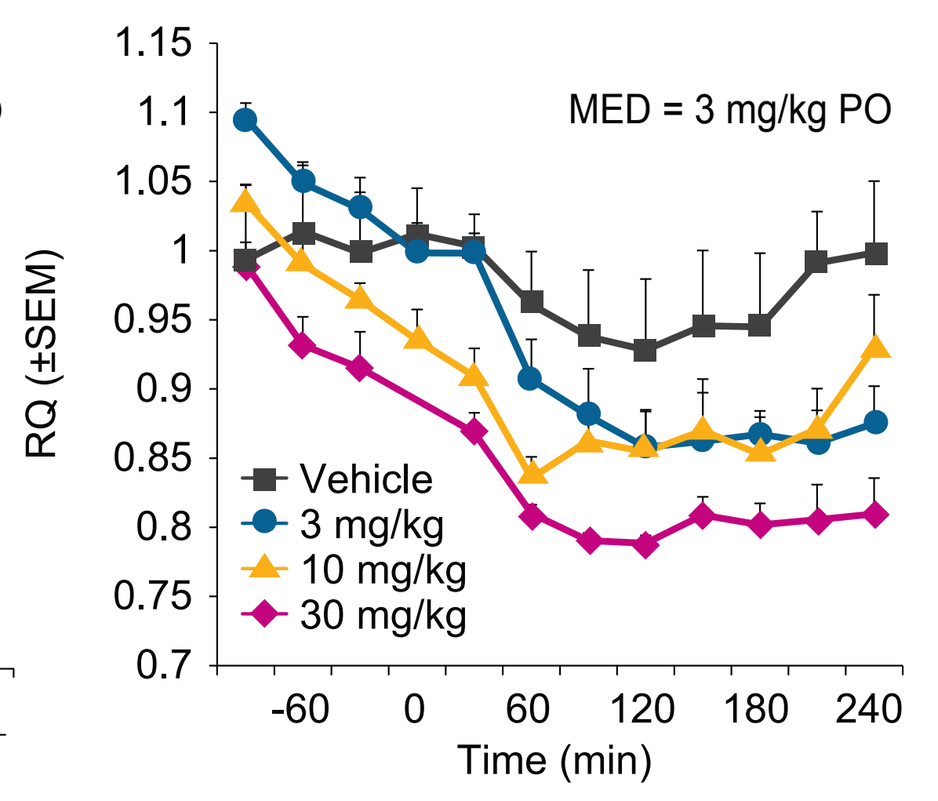
Target Engagement in Muscle
(Respiratory quotient)
MED = 3 mg/kg PO
RQ (±SEM)
Vehicle
3 mg/kg
10 mg/kg
30 mg/kg
Time (min)

FIGURE 7: Data Summary of ND-630 in High Fat and High Sucrose DIO Rat Studies

- Nimbus' allosteric inhibitors show promise for diabetes, metabolic disease, and fatty liver disease.

Endpoints	◆ 28-day rat DIO study high sucrose diet with 4w diet run-in	▲ 14-day rat DIO study high fat diet with 4w diet run-in
Body weight	↓	↓
Food intake	-	-
Hepatic triglycerides	↓	↓
Hepatic cholesterol	-	↓
Insulin Sensitivity (oGTT)	↑	↑
Plasma leptin	↓	↓
Plasma triglycerides	↓	-
Plasma free fatty acids	↓	nd
Plasma cholesterol	↓	-
Plasma ketone bodies	↑	nd
Plasma insulin	-	↓
Plasma glucose	-	-

FIGURE 8: Weight Neutral Profile; Well Tolerated. Reduction in Weight Gain at High Dose ◆

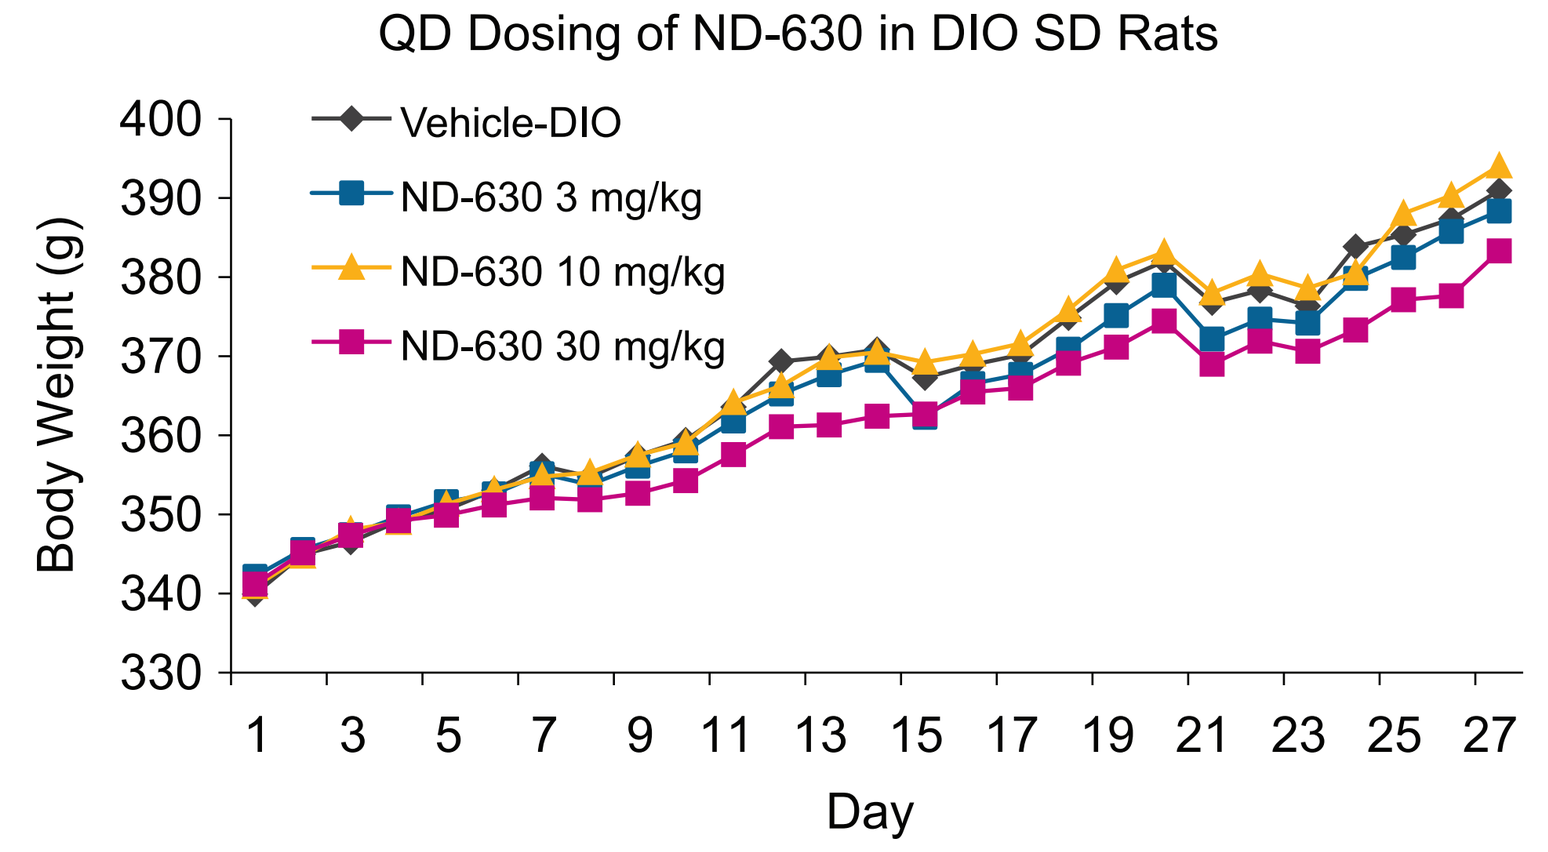
QD Dosing of ND-630 in DIO SD Rats
Body Weight (g)
Vehicle-DIO
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg
Day

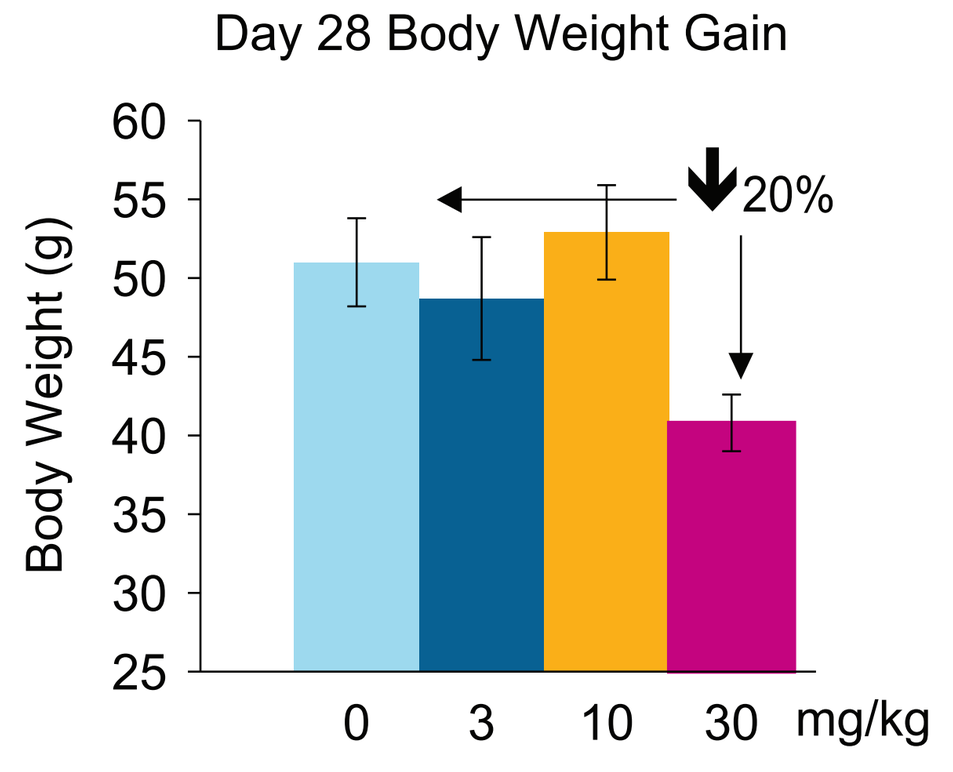
Day 28 Body Weight Gain
Body Weight (g)
20%
0 3 10 30 mg/kg

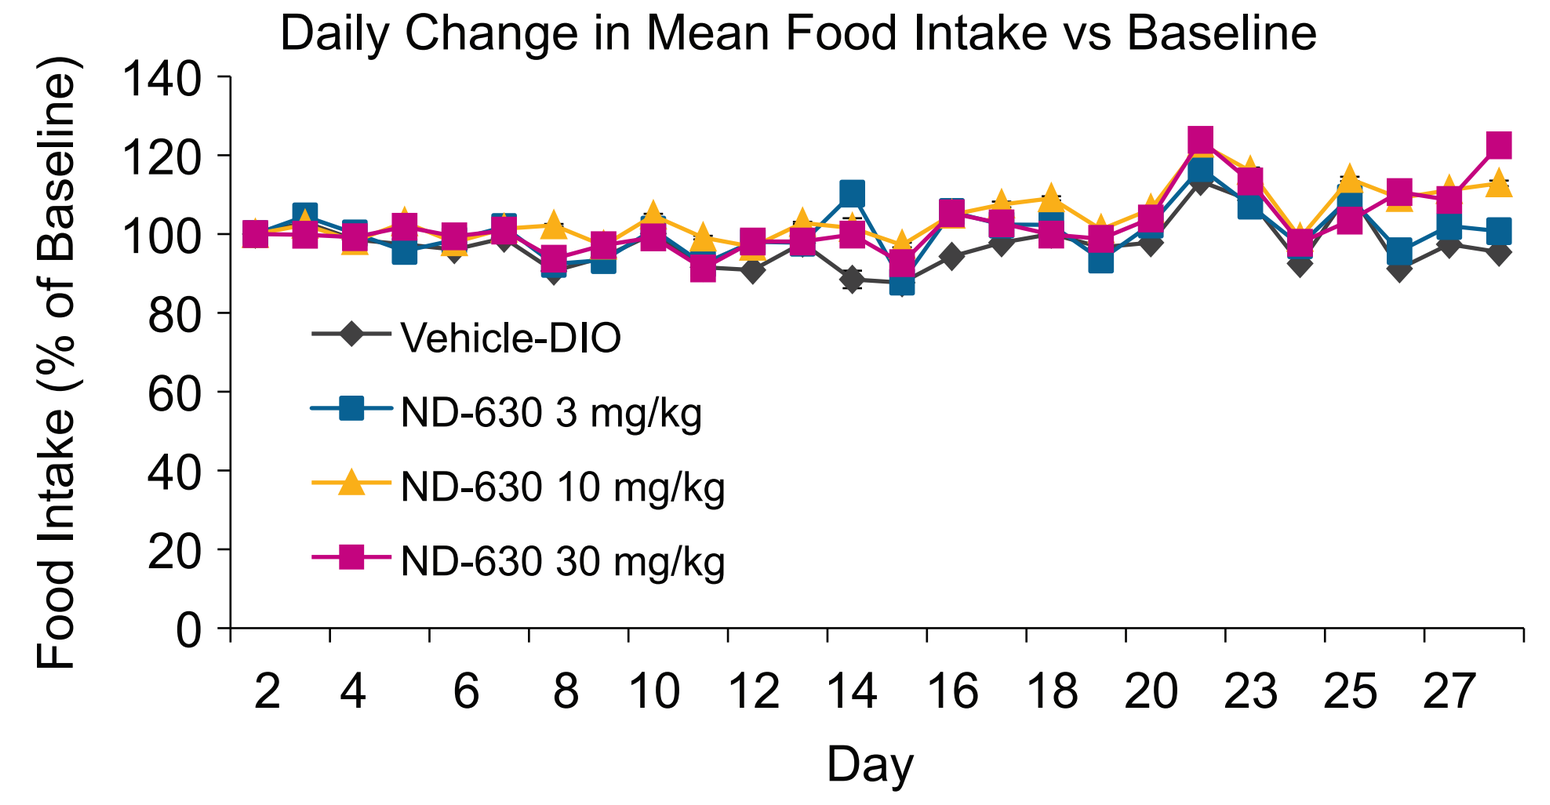
Daily Change in Mean Food Intake vs Baseline
Food Intake (% of Baseline)
Vehicle-DIO
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg
Day

- Weight neutral, with weight gain reduction at high dose.
- No change in food intake.
- No differences in fed blood glucose.
- Animals healthy.

FIGURE 9: Reduction in Whole Body Fat at All Doses ◆

Dose	Plasma Leptin (ng/ml) Prebleed*	Day 14*	Day 20**	Day 28**
0 mg/kg	6.72 ± 0.56	7.65 ± 0.64	9.00 ± 0.98	8.46 ± 0.64
3 mg/kg	6.63 ± 0.53	6.65 ± 0.67	8.01 ± 1.17	7.71 ± 1.09
10 mg/kg	6.66 ± 0.59	6.23 ± 1.01	5.42 ± 0.55	6.75 ± 0.78
30 mg/kg	6.42 ± 0.73	5.42 ± 0.52	4.85 ± 0.74	6.13 ± 0.89
Chow-fed Controls	4.20 ± 0.42	4.86 ± 0.56	5.34 ± 0.77	6.62 ± 0.81

- Evidence of decreased adiposity, resulting in impact on insulin sensitivity.

* Data are mean values for n=14 animals per group ± SEM
** Data are mean values for n=8 animals per group ± SEM

FIGURE 10: Improvement in Insulin Sensitivity ◆

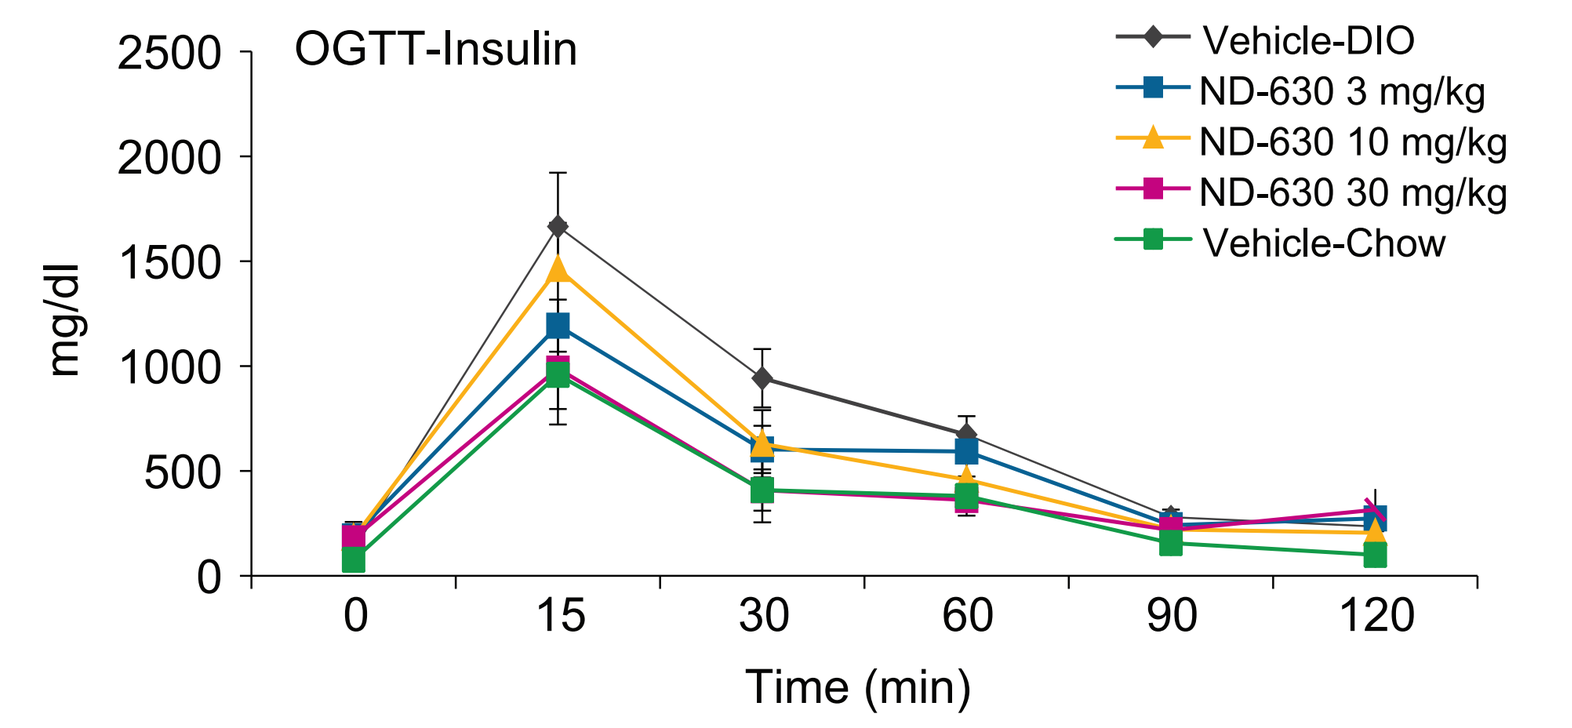
OGTT-Insulin
mg/dl
Vehicle-DIO
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg
Vehicle-Chow
Time (min)

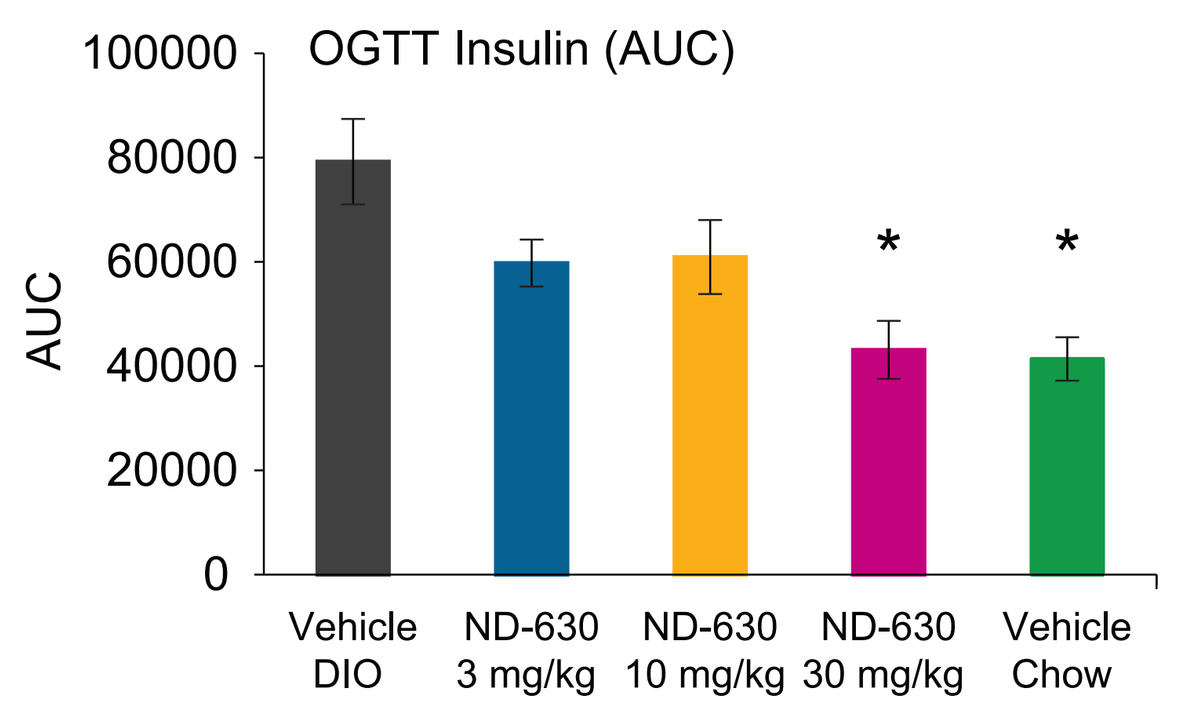
OGTT Insulin (AUC)
AUC
Vehicle DIO
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg
Vehicle Chow

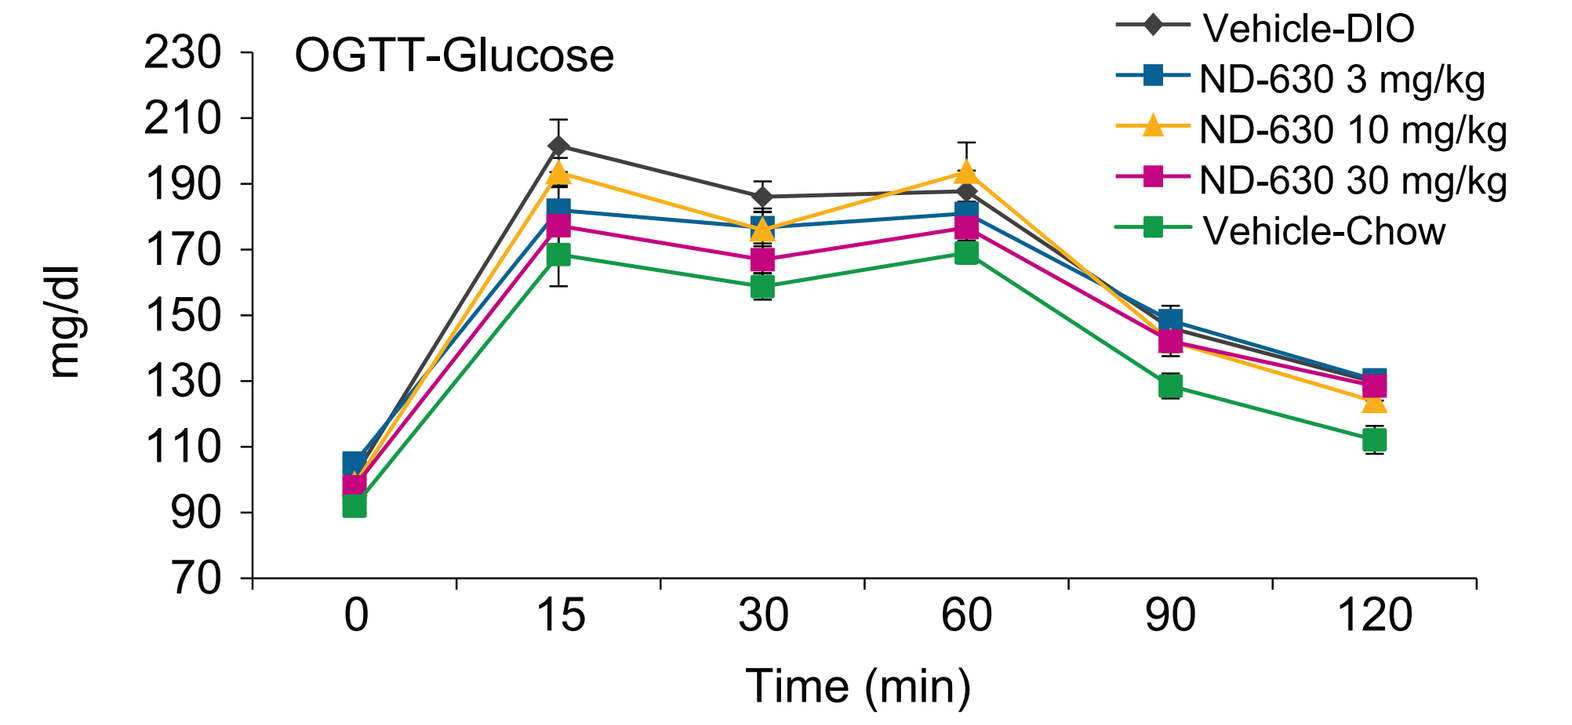
OGTT-Glucose
mg/dl
Vehicle-DIO
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg
Vehicle-Chow
Time (min)

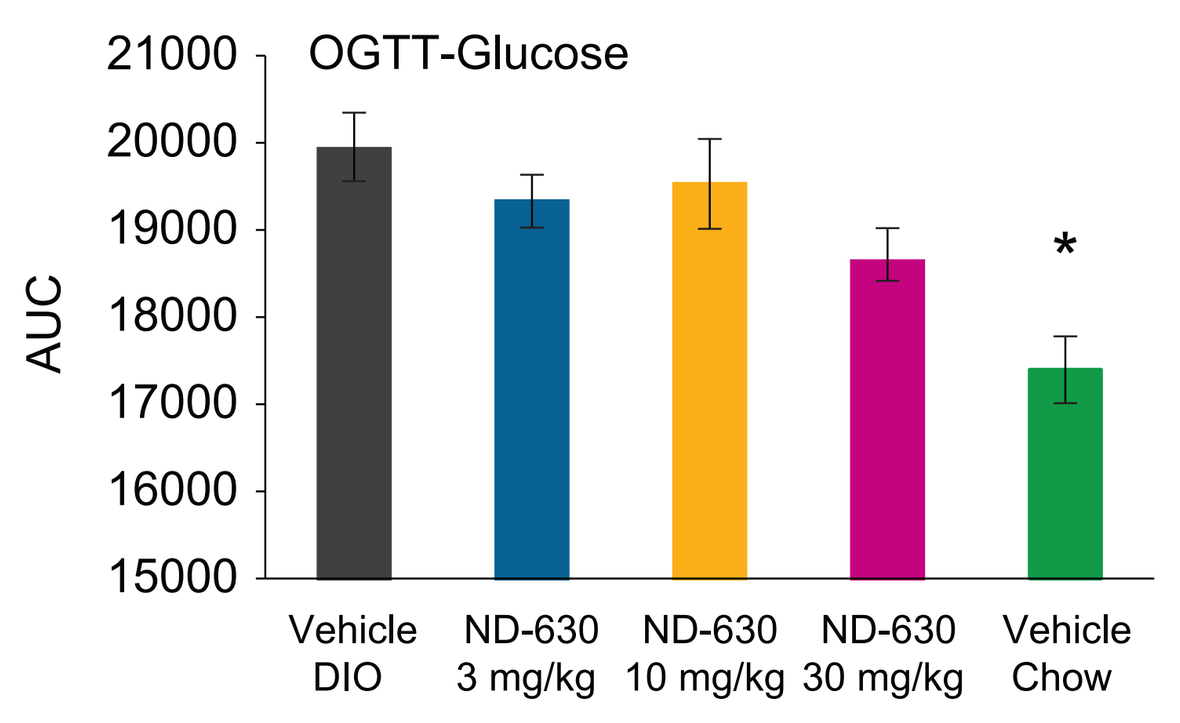
OGTT-Glucose
AUC
Vehicle DIO
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg
Vehicle Chow

FIGURE 11: Normalization of Hepatic Triglycerides and Reduced Hepatic Cholesterol ▲

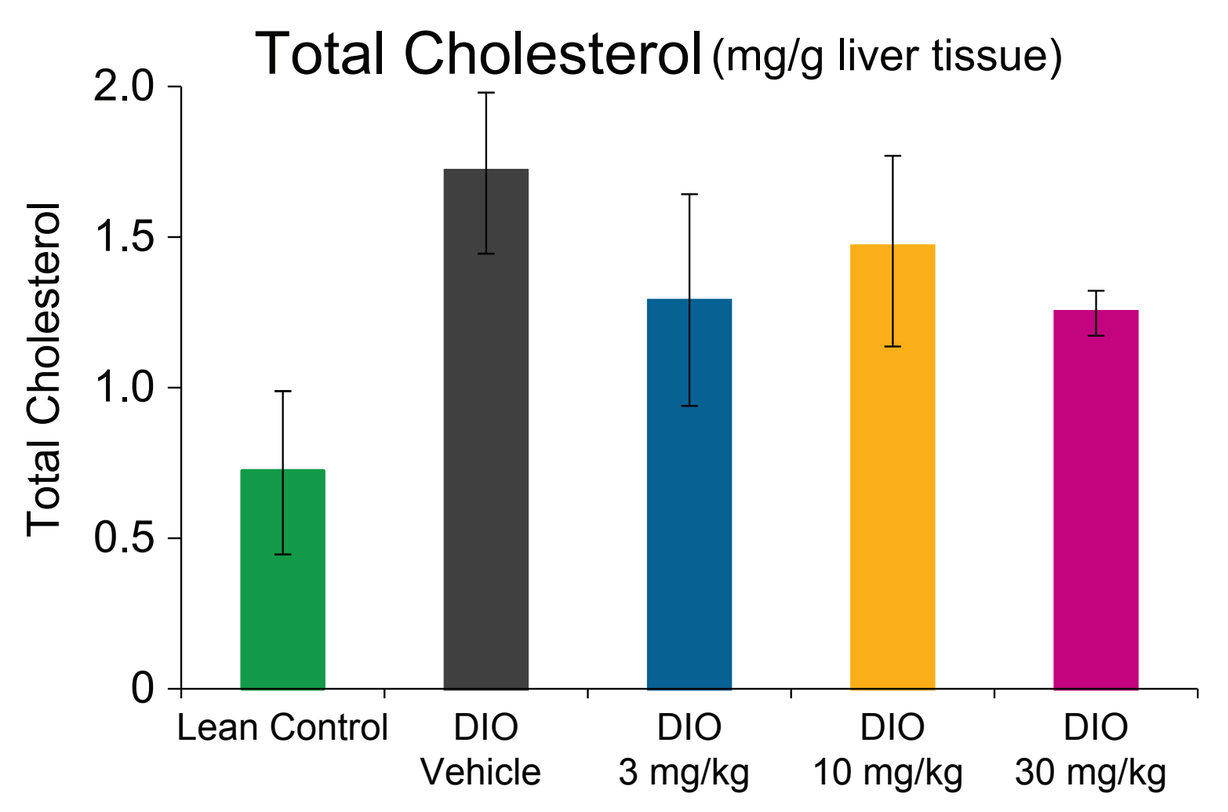
Total Cholesterol (mg/g liver tissue)
Total Cholesterol
Lean Control
DIO Vehicle
DIO 3 mg/kg
DIO 10 mg/kg
DIO 30 mg/kg

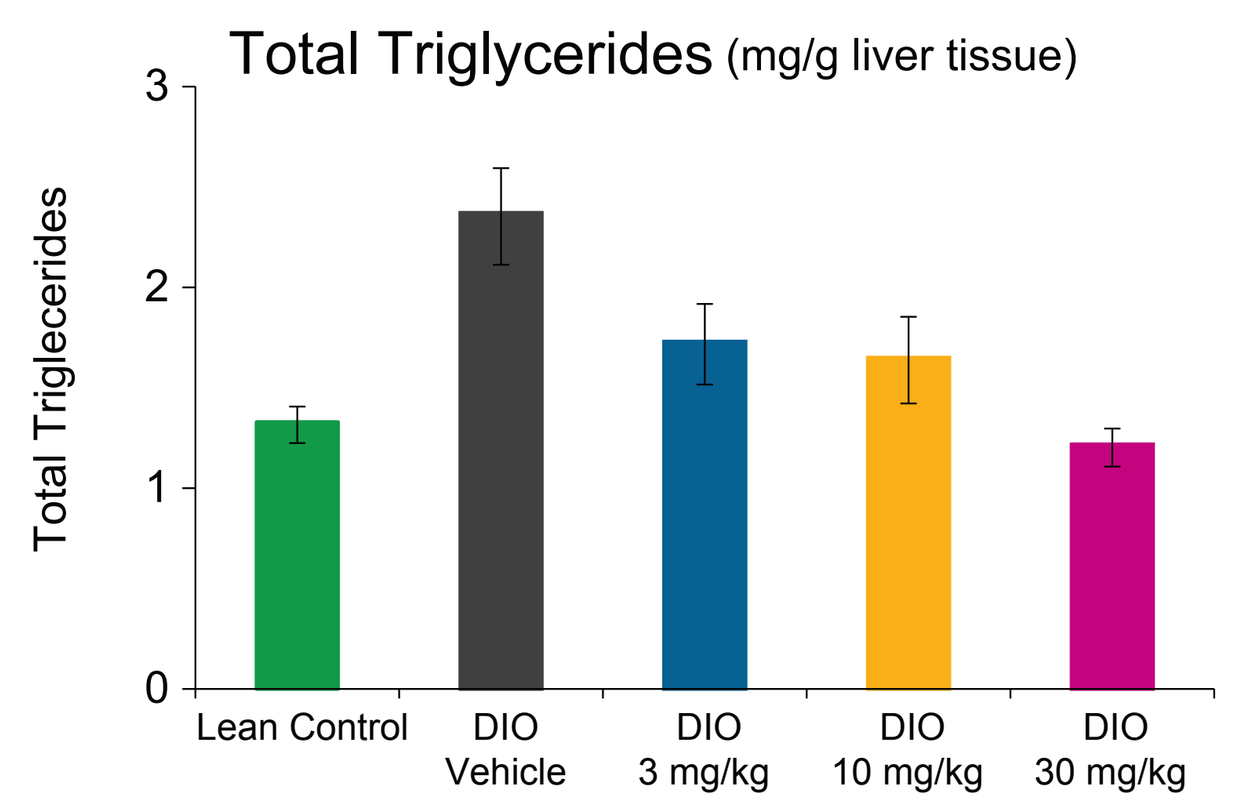
Total Triglycerides (mg/g liver tissue)
Total Triglecerides
Lean Control
DIO Vehicle
DIO 3 mg/kg
DIO 10 mg/kg
DIO 30 mg/kg

FIGURE 12: Dose Dependent Decrease of Plasma Triglycerides and Free Fatty Acids ◆

* Data are mean values for n=8 animals per group ± SEM

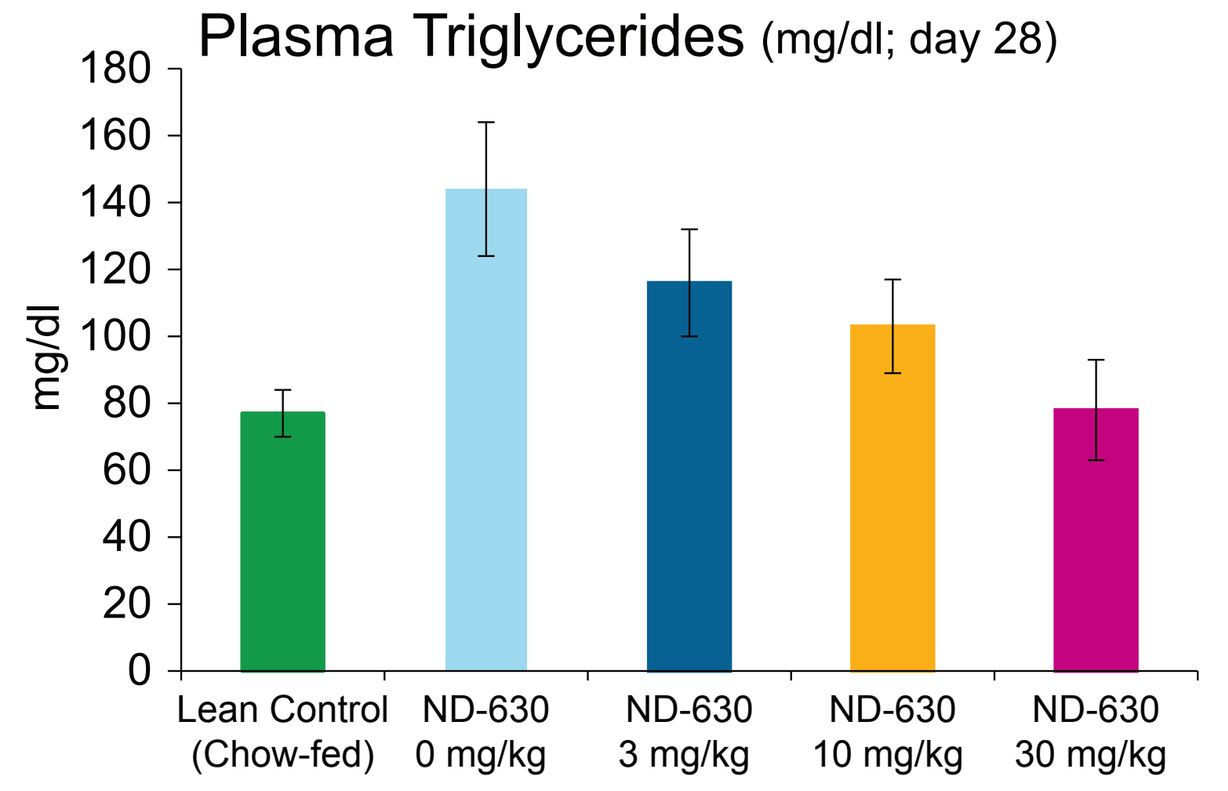
Plasma Triglycerides (mg/dl; day 28)
mg/dl
Lean Control (Chow-fed)
ND-630 0 mg/kg
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg

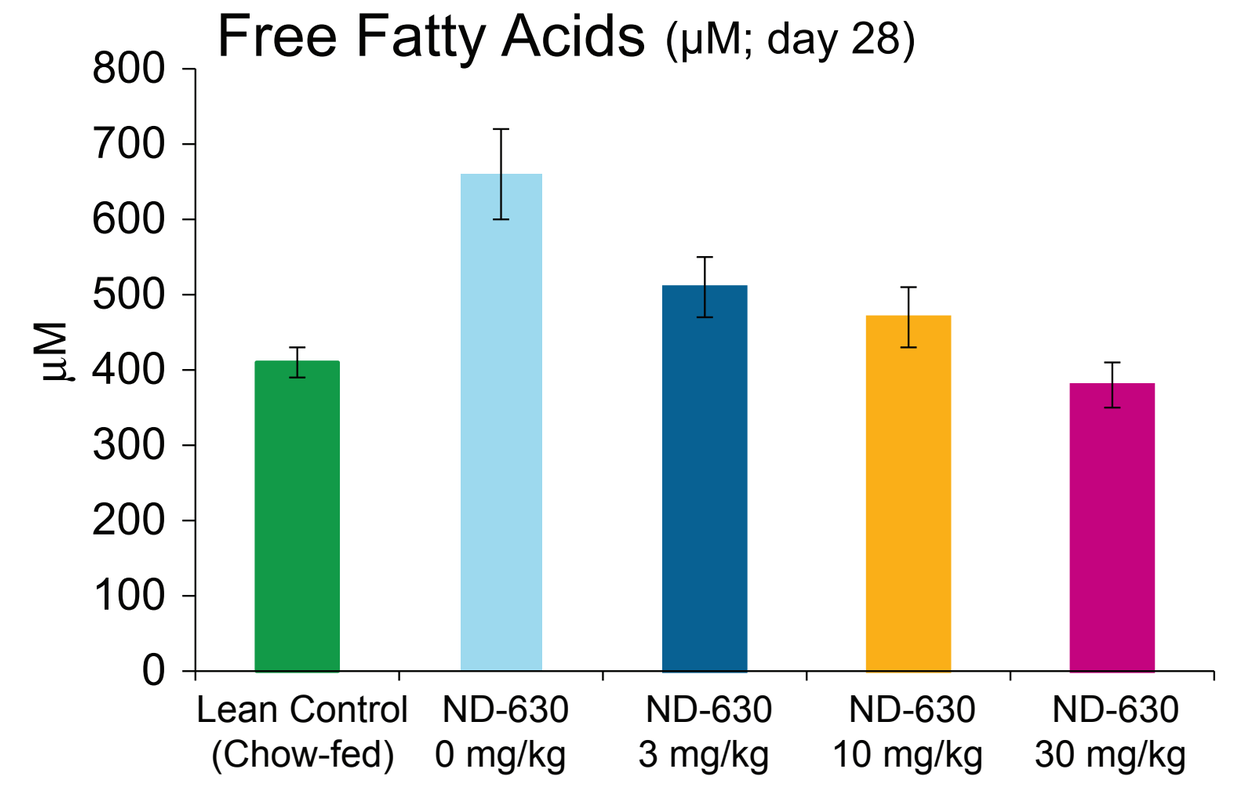
Free Fatty Acids (µM; day 28)
µM
Lean Control (Chow-fed)
ND-630 0 mg/kg
ND-630 3 mg/kg
ND-630 10 mg/kg
ND-630 30 mg/kg

FIGURE 13: Dose Dependent Decrease of Plasma Cholesterol ◆

Dose	Plasma Cholesterol (mg/dl) Prebleed*	Day 7*	Day 14*	Day 20**	Day 28**
0 mg/kg	100 ± 3	106 ± 3	110 ± 6	118 ± 5	107 ± 4
3 mg/kg	96 ± 2	94 ± 3	96 ± 3	101 ± 4	102 ± 4
10 mg/kg	96 ± 3	82 ± 3	87 ± 3	86 ± 4	91 ± 3
30 mg/kg	99 ± 3	78 ± 3	78 ± 2	74 ± 3	90 ± 3
Chow-fed Controls	109 ± 2	107 ± 3	107 ± 3	111 ± 6	103 ± 3

- ND-630 Reduces both plasma cholesterol & plasma triglycerides.

* Data are mean values for n=14 animals per group ± SEM
** Data are mean values for n=8 animals per group ± SEM

FIGURE 14: Increase Noted in Total Ketones ◆

* Data are mean values for n=8 animals per group ± SEM

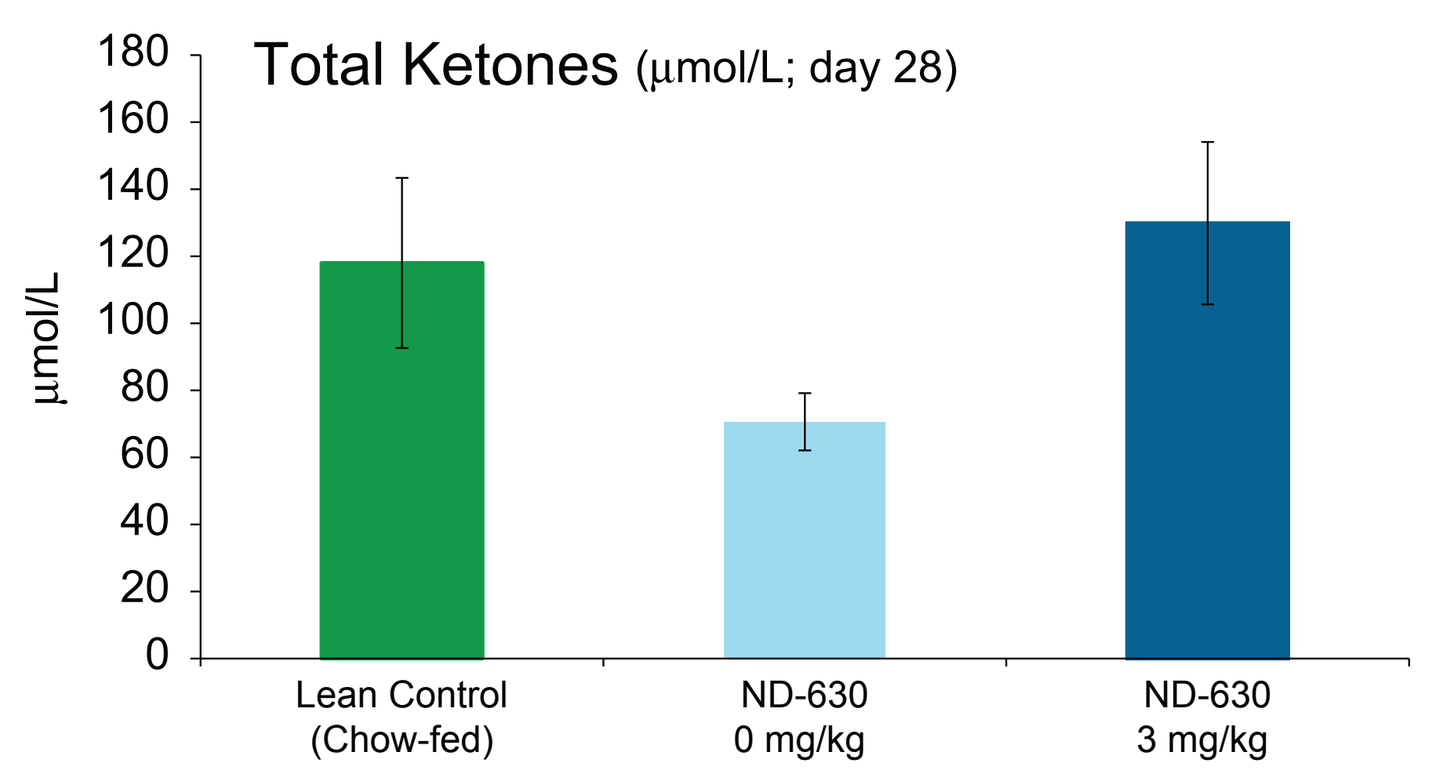
Total Ketones (µmol/L; day 28)
µmol/L
Lean Control (Chow-fed)
ND-630 0 mg/kg
ND-630 3 mg/kg

SUMMARY

- A structure-based virtual screen and drug design approach utilizing WaterMap was used to identify allosteric inhibitors of ACC that uniquely bind to the BC domain
- Inhibitors were successfully optimized for excellent potency, drug-like properties and *in vivo* efficacy in 16 months
- ND-630 demonstrated in-vivo proof of concept in pharmacologically relevant models of target engagement and efficacy (rat FASyn ED_{50} = 0.14 mpk, PO & rat RQ MED = 3 mg/kg, PO, MED 3 mg/kg DIO model)

ONGOING STUDIES

- Evaluation of ND-630 in ZDF diabetic rat model
- Evaluation of ND-630 in Hamster Lipid-lowering model

Nimbus Discovery - 25 First Street, Suite 404 - Cambridge, MA 02141 USA - Phone: (617) 588-2655 - Fax: (617) 231-0652 - Email:info@nimbusdiscovery.com - Web:www.nimbusdiscovery.com